TPS8059

Pomalidomide and Dexamethasone (Pom-Dex) With or Without Daratumumab (DARA) in Patients (Pts) With Relapsed or Refractory Multiple Myeloma (RRMM): a Multicenter, Randomized, Phase 3 Study (APOLLO)

Pieter Sonneveld,[1] Evangelos Terpos,[2] Meletios A. Dimopoulos,[2] Jon Ukropec,[3] Elena Smith,[4] Nienke Houkes,[5] Jordan M. Schecter,[6] Efstathios Kastritis[2]

[1]Department of Hematology, Erasmus MC, Rotterdam, The Netherlands; [2]Department of Clinical Therapeutics, National and Kapodistrian University of Athens, School of Medicine, "Alexandra" General Hospital, Athens, Greece; [3]Janssen Scientific Affairs, Horsham, PA, USA; [4]Janssen Research & Development, High Wycombe, UK; [5]Janssen Research & Development, Leiden, The Netherlands; [6]Janssen Research & Development, Raritan, NJ, USA.

Email: p.sonneveld@erasmusmc.nl

INTRODUCTION

- Daratumumab (DARA) is a human CD38-targeting monoclonal antibody with on-tumor and immunomodulatory mechanisms of action (**Figure 1**)
 - On-tumor activities include: complement-dependent cytotoxicity,[1] antibody-dependent cellular cytotoxicity,[1] antibody-dependent cellular phagocytosis,[2] apoptosis,[3] and direct enzymatic inhibition[4]
 - Immunomodulatory activities include: inducing lysis of myeloid-derived suppressor cells, regulatory B cells, and a subpopulation of regulatory T cells ($CD4^{+}CD25^{+}CD127^{dim}$) to promote T-cell activity[5]

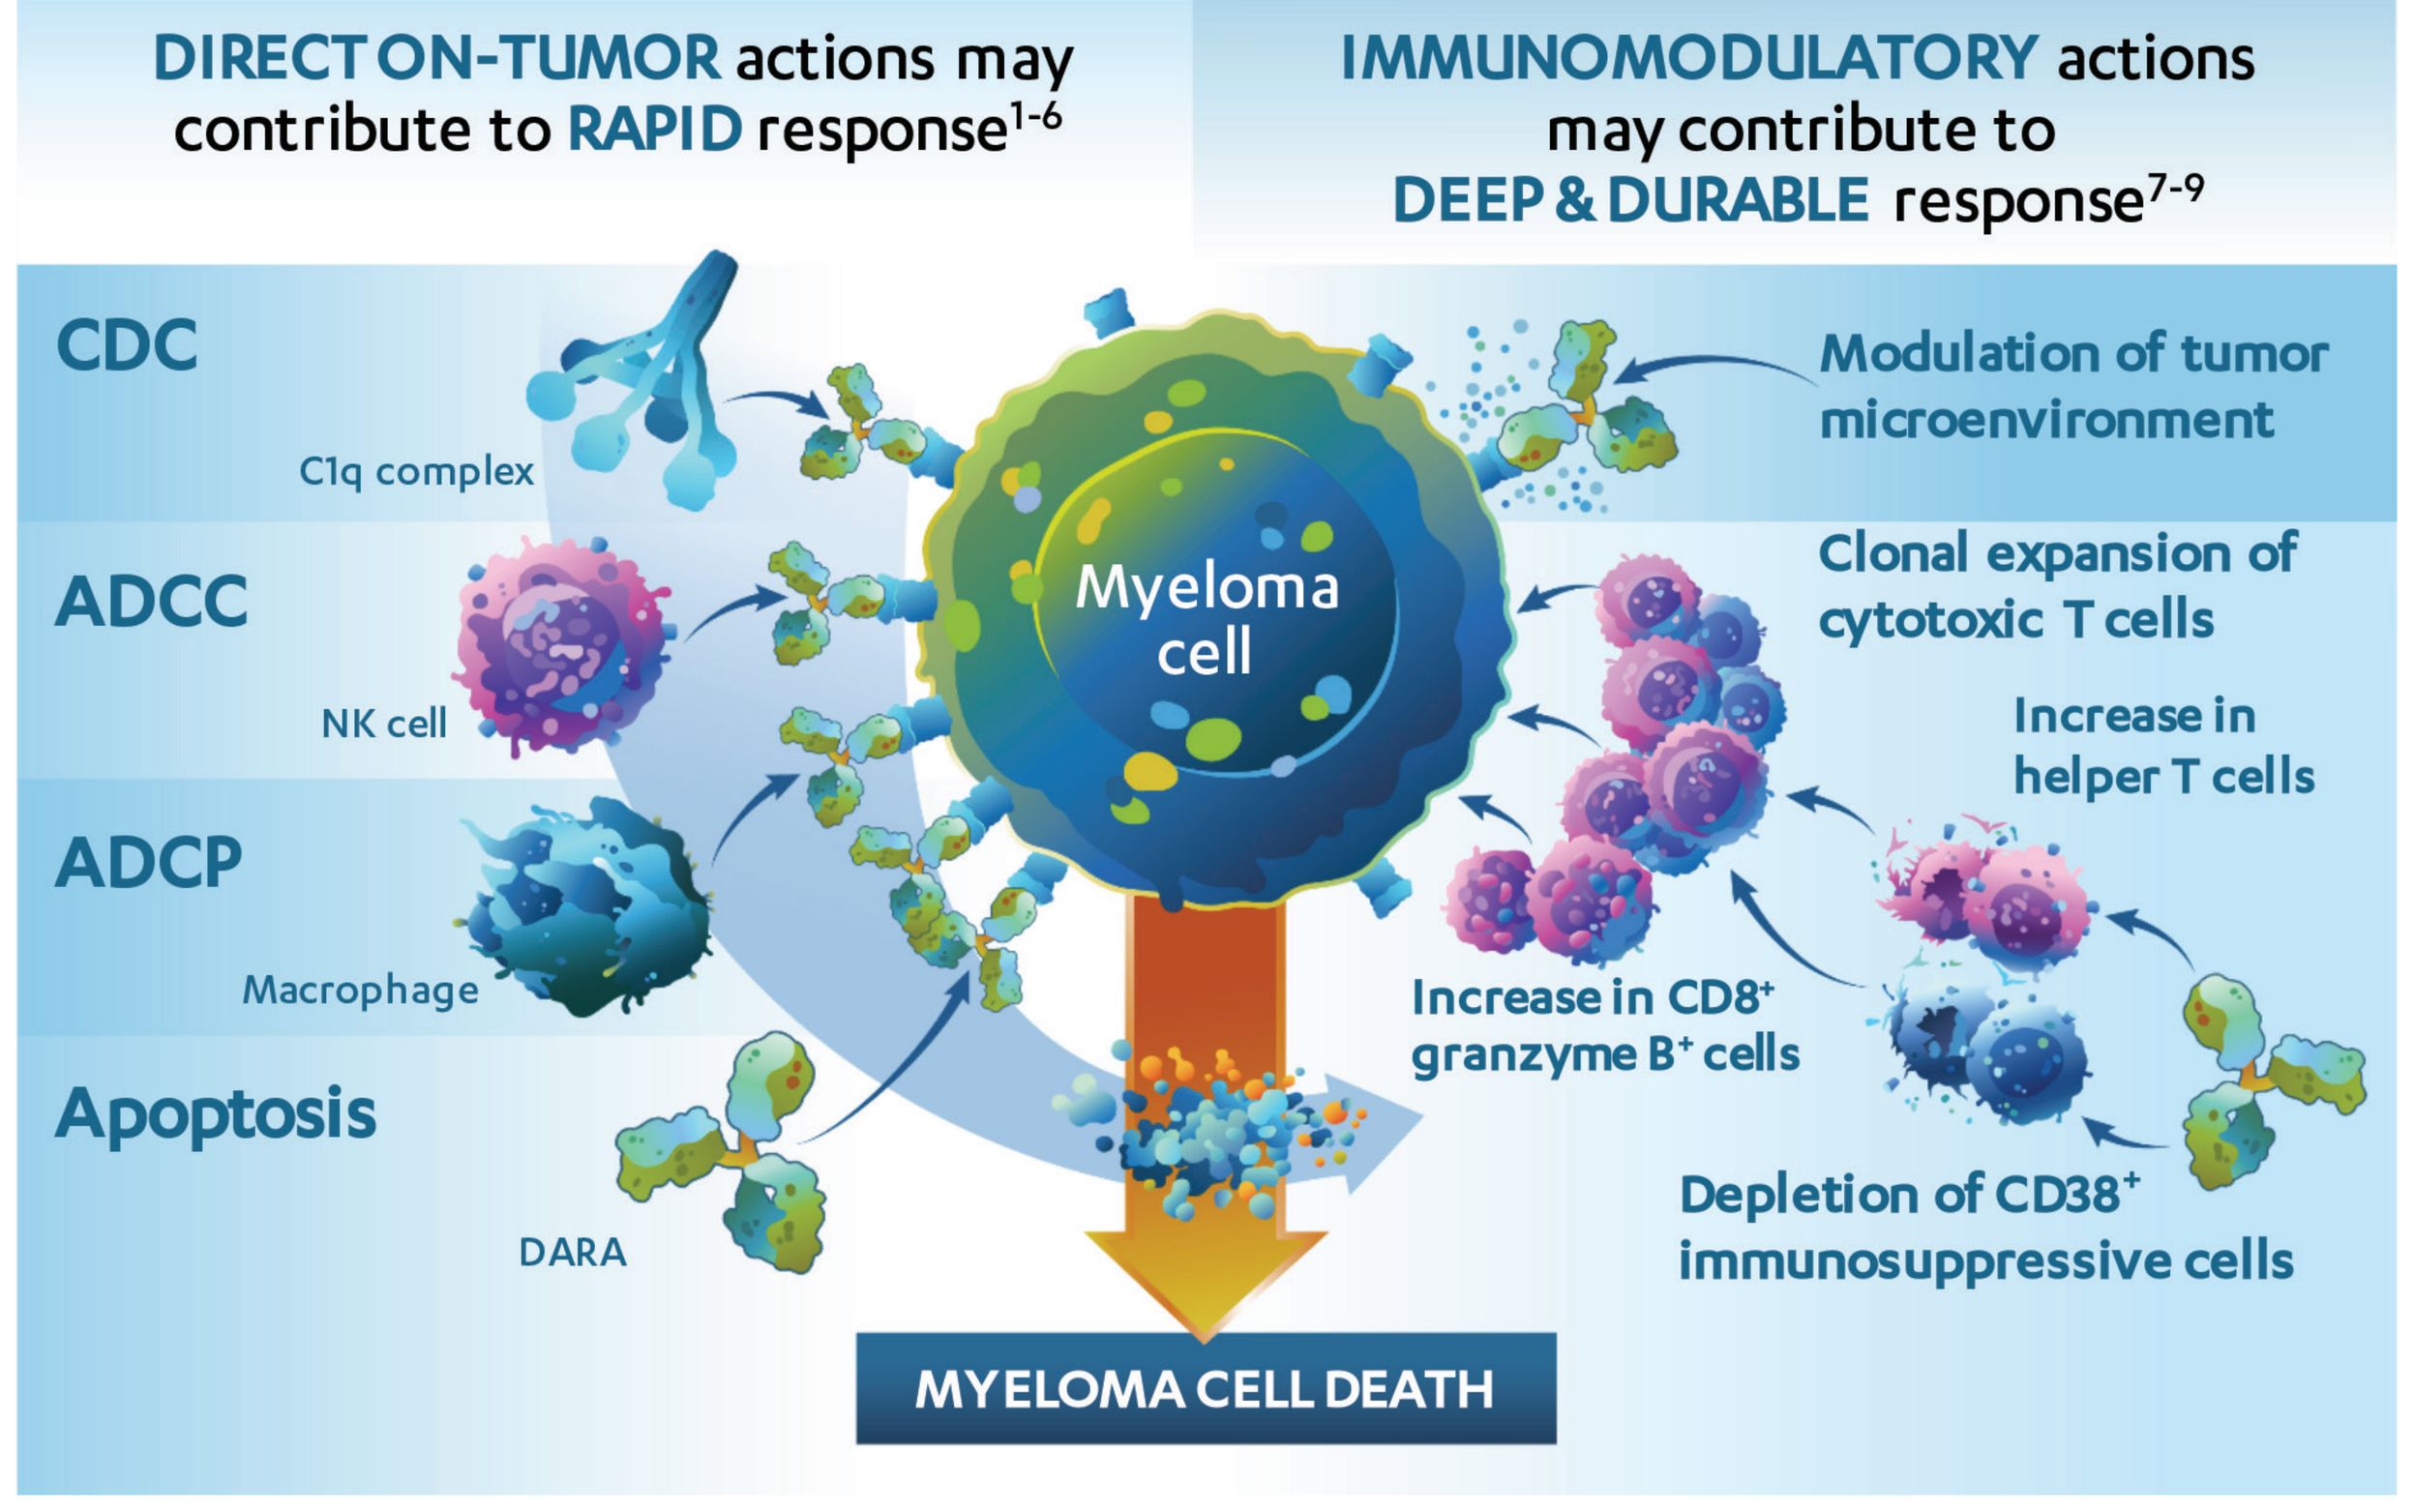
DIRECT ON-TUMOR actions may contribute to RAPID response1-6
IMMUNOMODULATORY actions may contribute to DEEP & DURABLE response7-9
CDC
C1q complex
ADCC
NK cell
ADCP
Macrophage
Apoptosis
DARA
Myeloma cell
Modulation of tumor microenvironment
Clonal expansion of cytotoxic T cells
Increase in helper T cells
Increase in CD8+ granzyme B+ cells
Depletion of CD38+ immunosuppressive cells
MYELOMA CELL DEATH

DARA, daratumumab; CDC, complement-dependent cytotoxicity; ADCC, antibody-dependent cellular cytotoxicity; NK, natural killer; ADCP, antibody-dependent cellular phagocytosis.

1. DARZALEX® (daratumumab) injection, for intravenous use [package insert]. Horsham, PA: Janssen Biotech, Inc.; 2018. 2. Liszewski MK, et al. *Adv Immunol*. 1996;61:201-283. 3. Debets JM, et al. *J Immunol*. 1988;141(4):1197-1201. 4. Overdijk MB, et al. *MAbs*. 2015;7(2):311-321. 5. Lokhorst HM, et al. *N Engl J Med*. 2015;373(13):1207-1219. 6. Plesner T, et al. Presented at: ASH; December 8-11, 2012; Atlanta, GA. Abstract 73. 7. Krejcik J, et al. *Blood*. 2016;128(3):384-394. 8. Adams H, et al. Presented at: ASH; December 3-6, 2016; San Diego, CA. Abstract 4521. 9. Chiu C, et al. Presented at: ASH; December 3-6, 2016; San Diego, CA. Abstract 4531.

Figure 1. DARA mechanism of action.

- Pomalidomide (Pom) is an analog of thalidomide that stimulates T cells and natural killer cells, inhibits proinflammatory cytokine production, demonstrates antiangiogenic activity, inhibits proliferation, and induces apoptosis of tumor cells[6]
- Pom in combination with dexamethasone (Pom-Dex) is approved for multiple myeloma (MM) patients who have received ≥2 prior therapies, including lenalidomide and a proteasome inhibitor (PI), and have demonstrated disease progression on or within 60 days of completion of the last therapy[7]
- In a phase 1b study of DARA in combination with standard of care regimens (MMY1001), DARA (administered intravenously [IV]) in combination with Pom-Dex demonstrated an overall response rate of 60%, median progression-free survival (PFS) of 8.8 months, and median overall survival of 17.5 months in heavily pre-treated relapsed or refractory MM (RRMM) patients[8]
 - On June 16, 2017, the US Food and Drug Administration approved DARA in combination with Pom-Dex for MM patients who received ≥2 prior therapies, including lenalidomide and a PI[9]
 - DARA (16 mg/kg IV) is also approved as monotherapy and in combination with lenalidomide and Dex or bortezomib and Dex for treatment of RRMM patients, and in combination with bortezomib, melphalan, and prednisone for the treatment of patients with newly diagnosed MM who are ineligible for autologous stem cell transplant[9]
- A subcutaneous formulation of DARA (DARA SC; 1,800 mg co-formulated with recombinant human hyaluronidase PH20 [rHuPH20; ENHANZE® drug delivery technology, Halozyme, Inc.]) demonstrated low rates of infusion-related reactions and response rates similar to DARA IV in a phase 1b study of RRMM patients[10]

OBJECTIVE

- To evaluate the efficacy and safety of DARA SC + Pom-Dex versus Pom-Dex alone in patients with RRMM who received ≥1 prior treatment with both lenalidomide and a PI

METHODS

Key Eligibility Criteria

- ≥18 years of age
- Eastern Cooperative Oncology Group performance status of ≤2
- Responded to prior treatment with lenalidomide and a PI and documented evidence of progressive disease
- Patients who received only 1 prior treatment must have demonstrated progressive disease ≤60 days after completing the lenalidomide-containing regimen
- Creatinine clearance ≥30 mL/min
- Measurable disease was defined as the following:
 - IgG MM: serum M-protein level ≥1.0 g/dL or urine M-protein level ≥200 mg/24 hours
 - IgA, IgD, IgE, IgM MM: serum M-protein level ≥0.5 g/dL or urine M-protein level ≥200 mg/24 hours
 - Light chain MM for patients without measurable disease in the serum or urine: serum Ig free light chain (FLC) ≥10 mg/dL and abnormal serum Ig kappa lambda FLC ratio
- No prior treatment with anti-CD38 therapies or Pom

Study Design

- APOLLO is a phase 3, randomized, open-label, multicenter study comparing DARA + Pom-Dex with Pom-Dex alone in patients with RRMM who received ≥1 prior treatment with both lenalidomide and a PI
- Approximately 302 patients will be randomized 1:1 to receive either DARA + Pom-Dex or Pom-Dex (**Figure 2**)
 - Safety evaluations will occur weekly during Cycles 1 and 2 (28-day cycles), every 2 weeks for Cycles 3 through 6, and on Day 1 of each cycle during Cycles ≥7
 - Disease evaluation will occur monthly

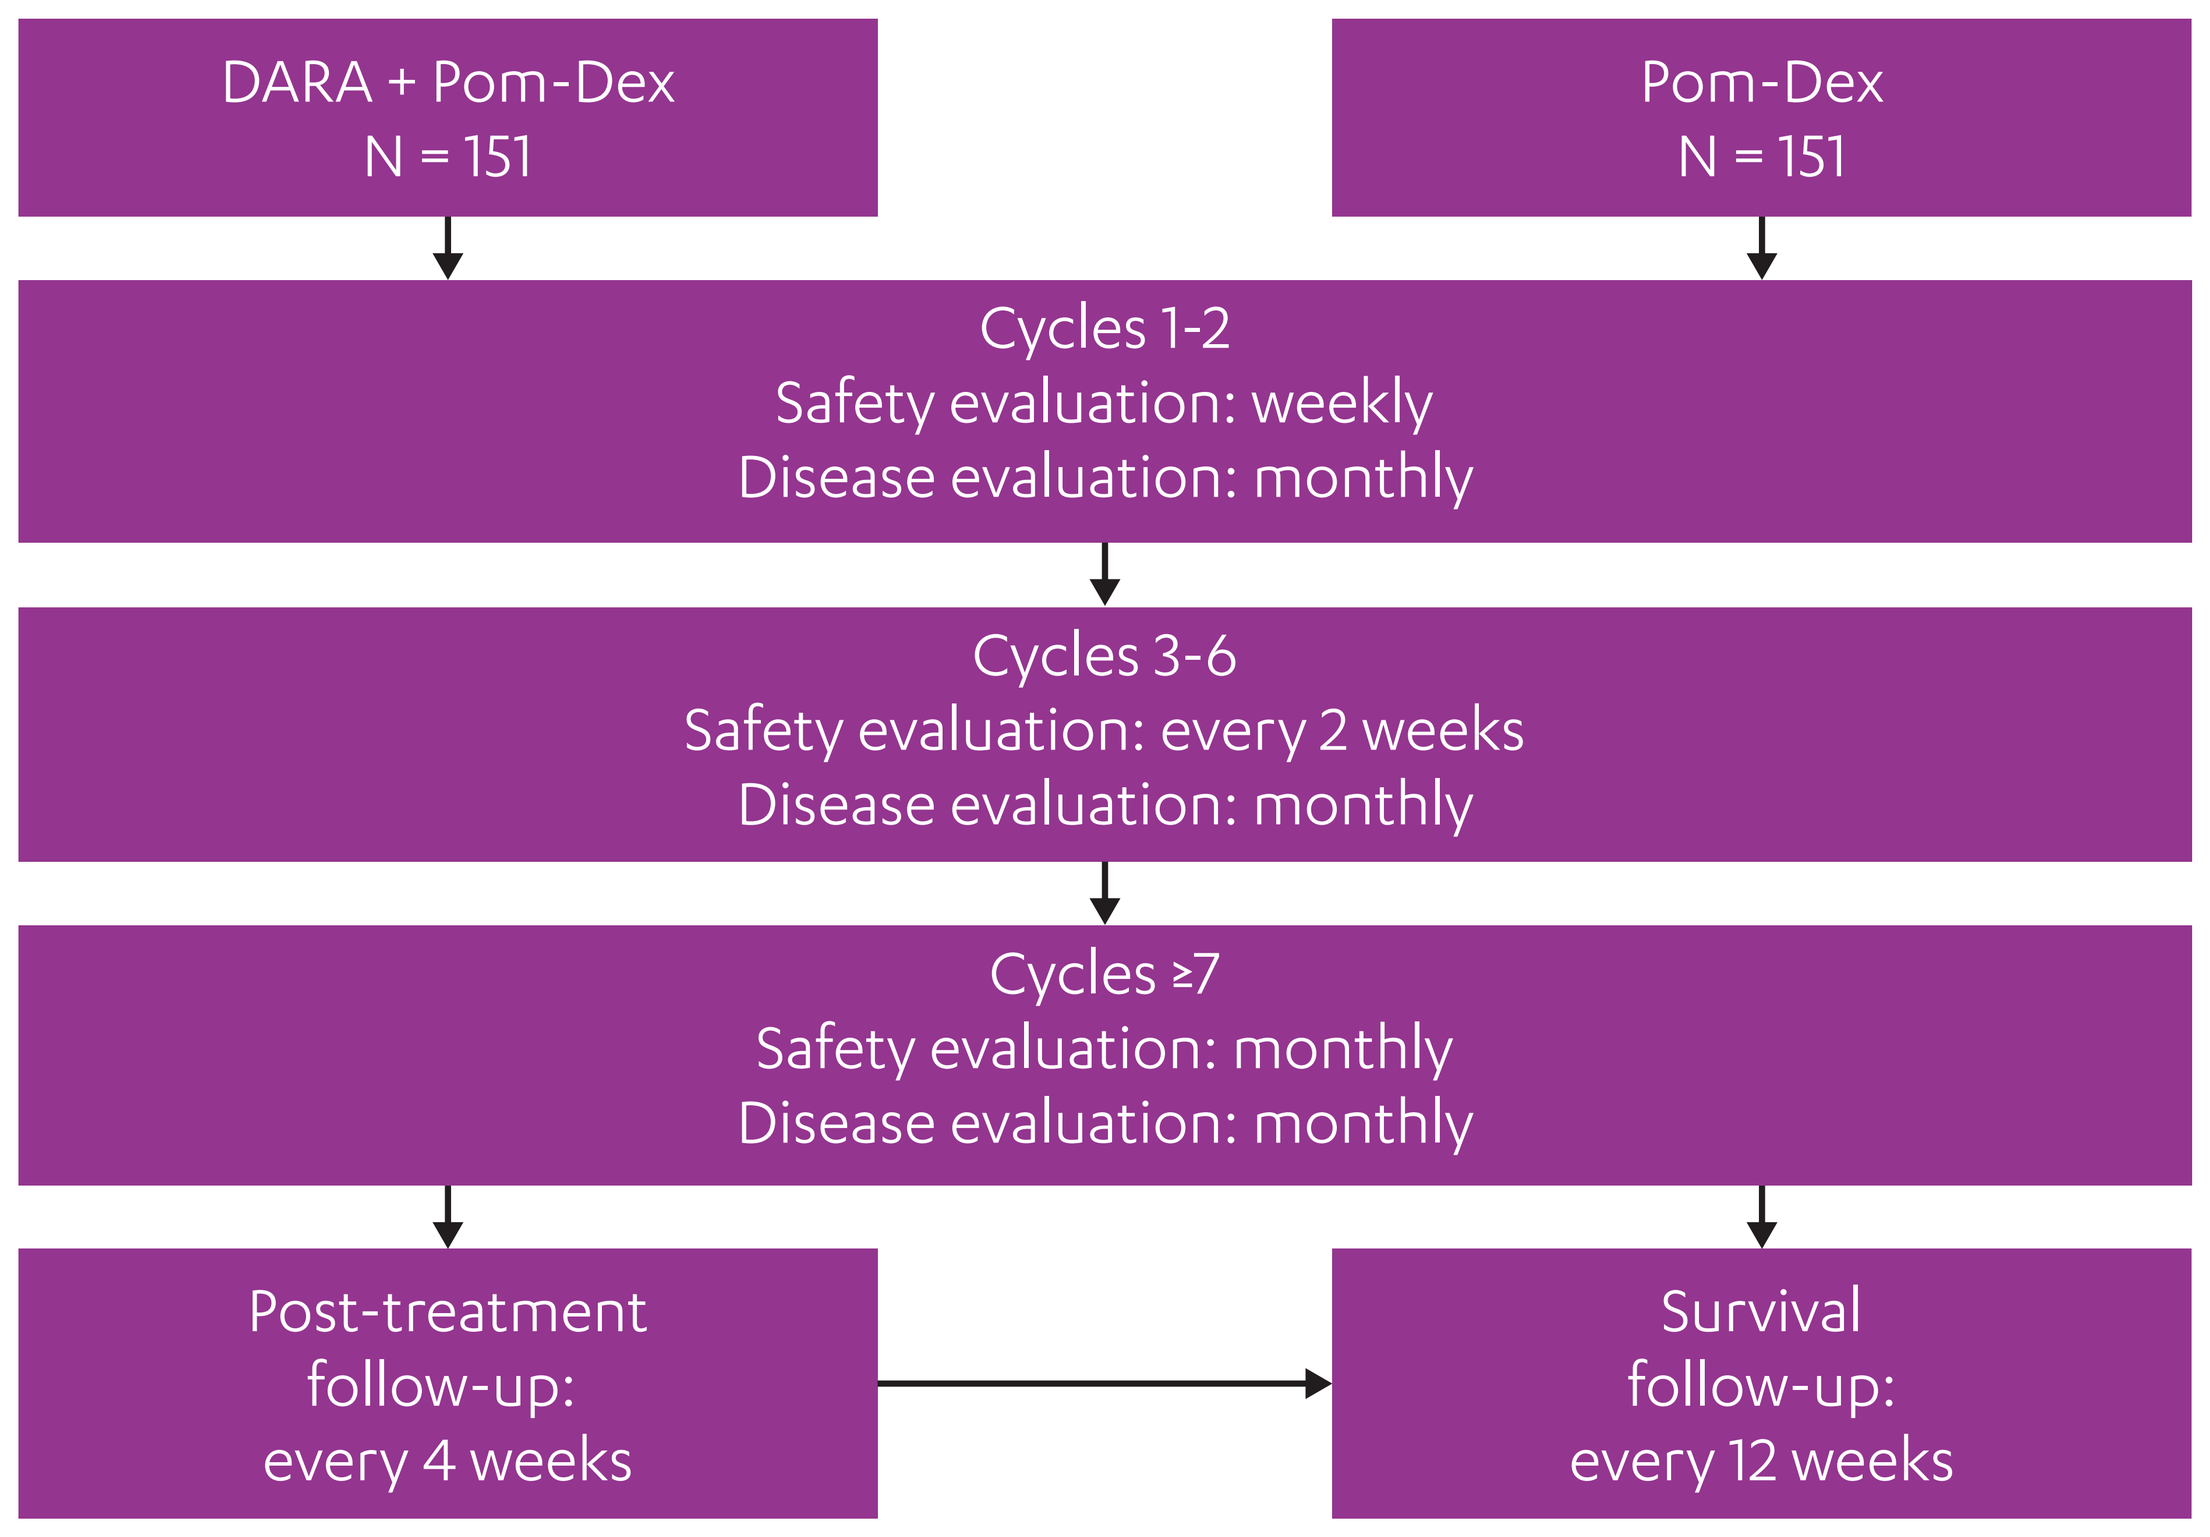
DARA + Pom-Dex
N = 151
Pom-Dex
N = 151
Cycles 1-2
Safety evaluation: weekly
Disease evaluation: monthly
Cycles 3-6
Safety evaluation: every 2 weeks
Disease evaluation: monthly
Cycles ≥7
Safety evaluation: monthly
Disease evaluation: monthly
Post-treatment follow-up: every 4 weeks
Survival follow-up: every 12 weeks

DARA + Pom-Dex, daratumumab plus pomalidomide and dexamethasone; Pom-Dex, pomalidomide and dexamethasone.

Figure 2. APOLLO study design.

- The dosing schedule consisting of 28-day cycles is summarized in **Figure 3**
 - DARA
 - Before a protocol amendment, patients received DARA 16 mg/kg IV once weekly for Cycles 1 and 2, every 2 weeks for Cycles 3 through 6, and every 4 weeks for Cycles ≥7
 - Following a protocol amendment, patients in the DARA + Pom-Dex cohort will receive DARA SC (1,800 mg co-formulated with rHuPH20) based on the same administration schedule
 - Pom-Dex
 - Pom (4 mg): daily for Days 1 through 21 in each cycle
 - Dex (40 mg; 20 mg for patients ≥75 years of age): weekly in each cycle

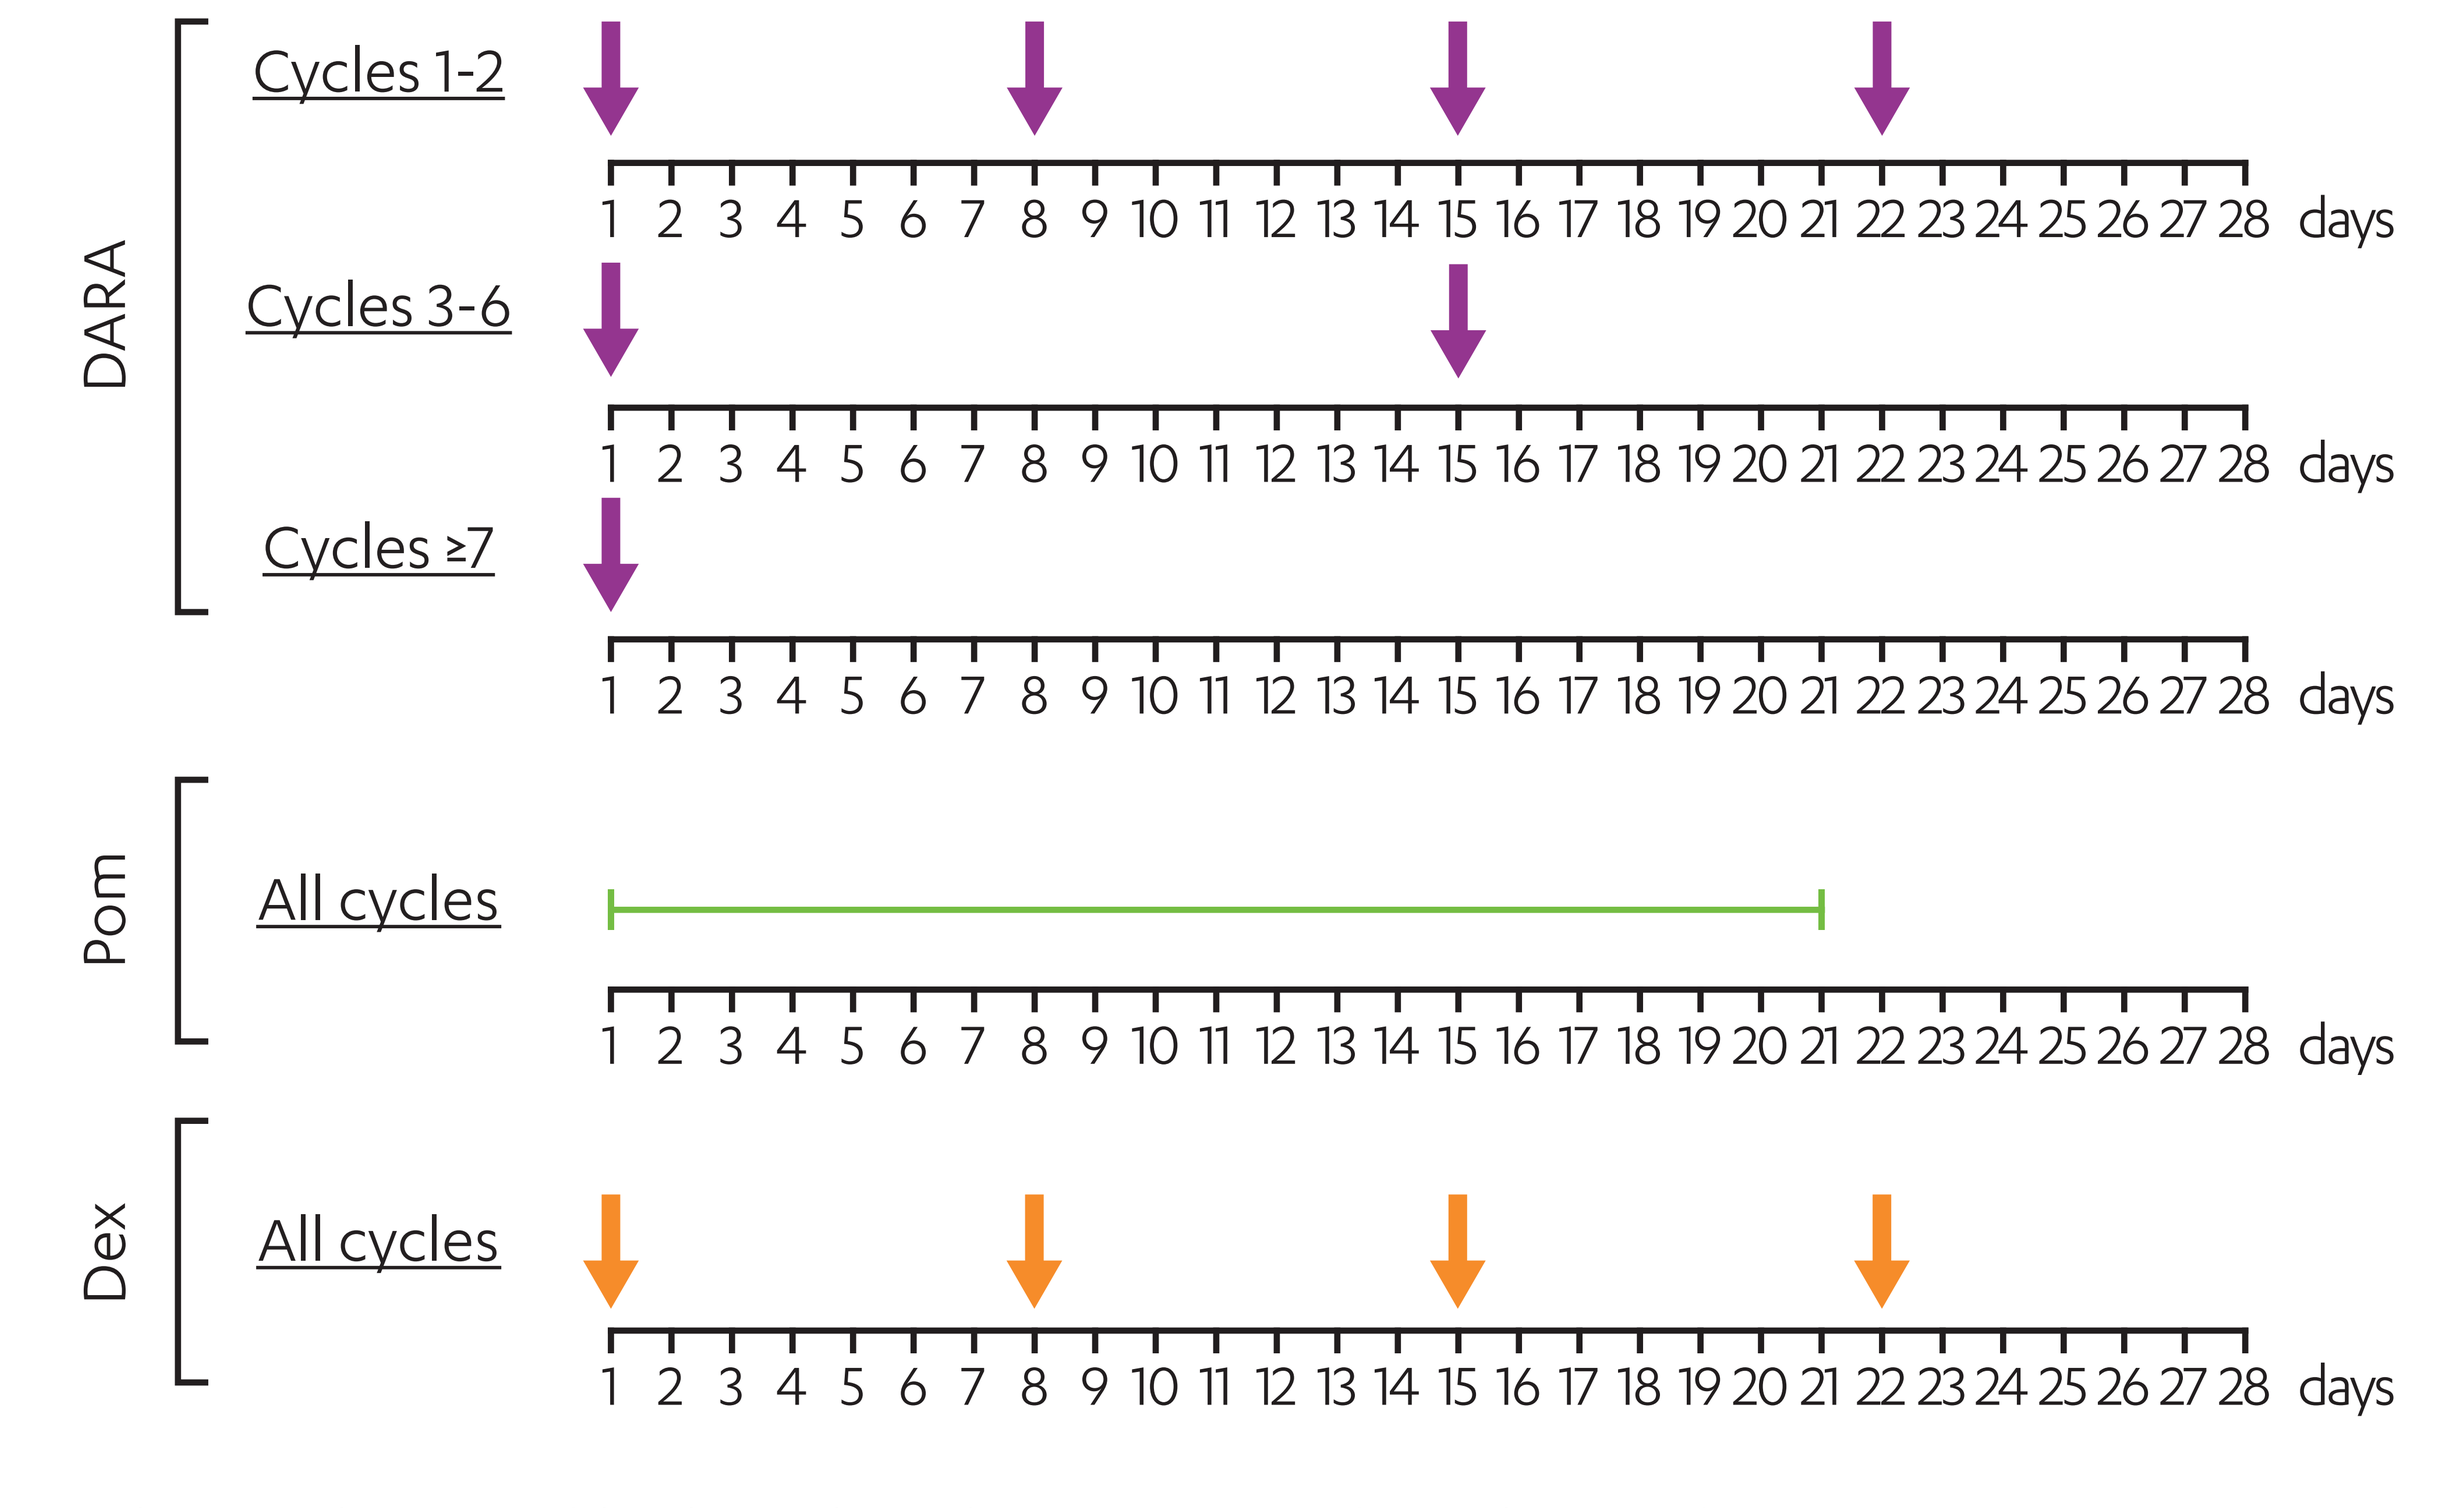
DARA
Cycles 1-2
Cycles 3-6
Cycles ≥7
Pom
All cycles
Dex
All cycles
1 2 3 4 5 6 7 8 9 10 11 12 13 14 15 16 17 18 19 20 21 22 23 24 25 26 27 28 days

DARA, daratumumab; Pom, pomalidomide; Dex, dexamethasone; IV, intravenously; SC, subcutaneously.
Arrows indicate when DARA and Dex are administered. DARA (16 mg/kg IV or 1,800 mg SC) is administered weekly during Cycles 1 and 2, every 2 weeks during Cycles 3 through 6, and every 4 weeks thereafter. Pom (4 mg orally) is administered on Days 1 through 21 of each cycle. Dex (40 mg; 20 mg for patients ≥75 years of age) is administered weekly. Patients will receive treatment until disease progression or unacceptable toxicity. Each cycle is 28 days.

Figure 3. APOLLO dosing schedule.

- To mitigate potential infusion-related reactions, all patients will receive pre-infusion medications, including Dex, acetaminophen, diphenhydramine, and an optional leukotriene inhibitor
- Patients with a higher risk of respiratory complications will receive post-infusion medications, including diphenhydramine, a short-acting β_2 adrenergic receptor agonist, and lung disease control medications
- A total of 302 patients are expected to enroll in sites that span 12 countries (**Figure 4**)

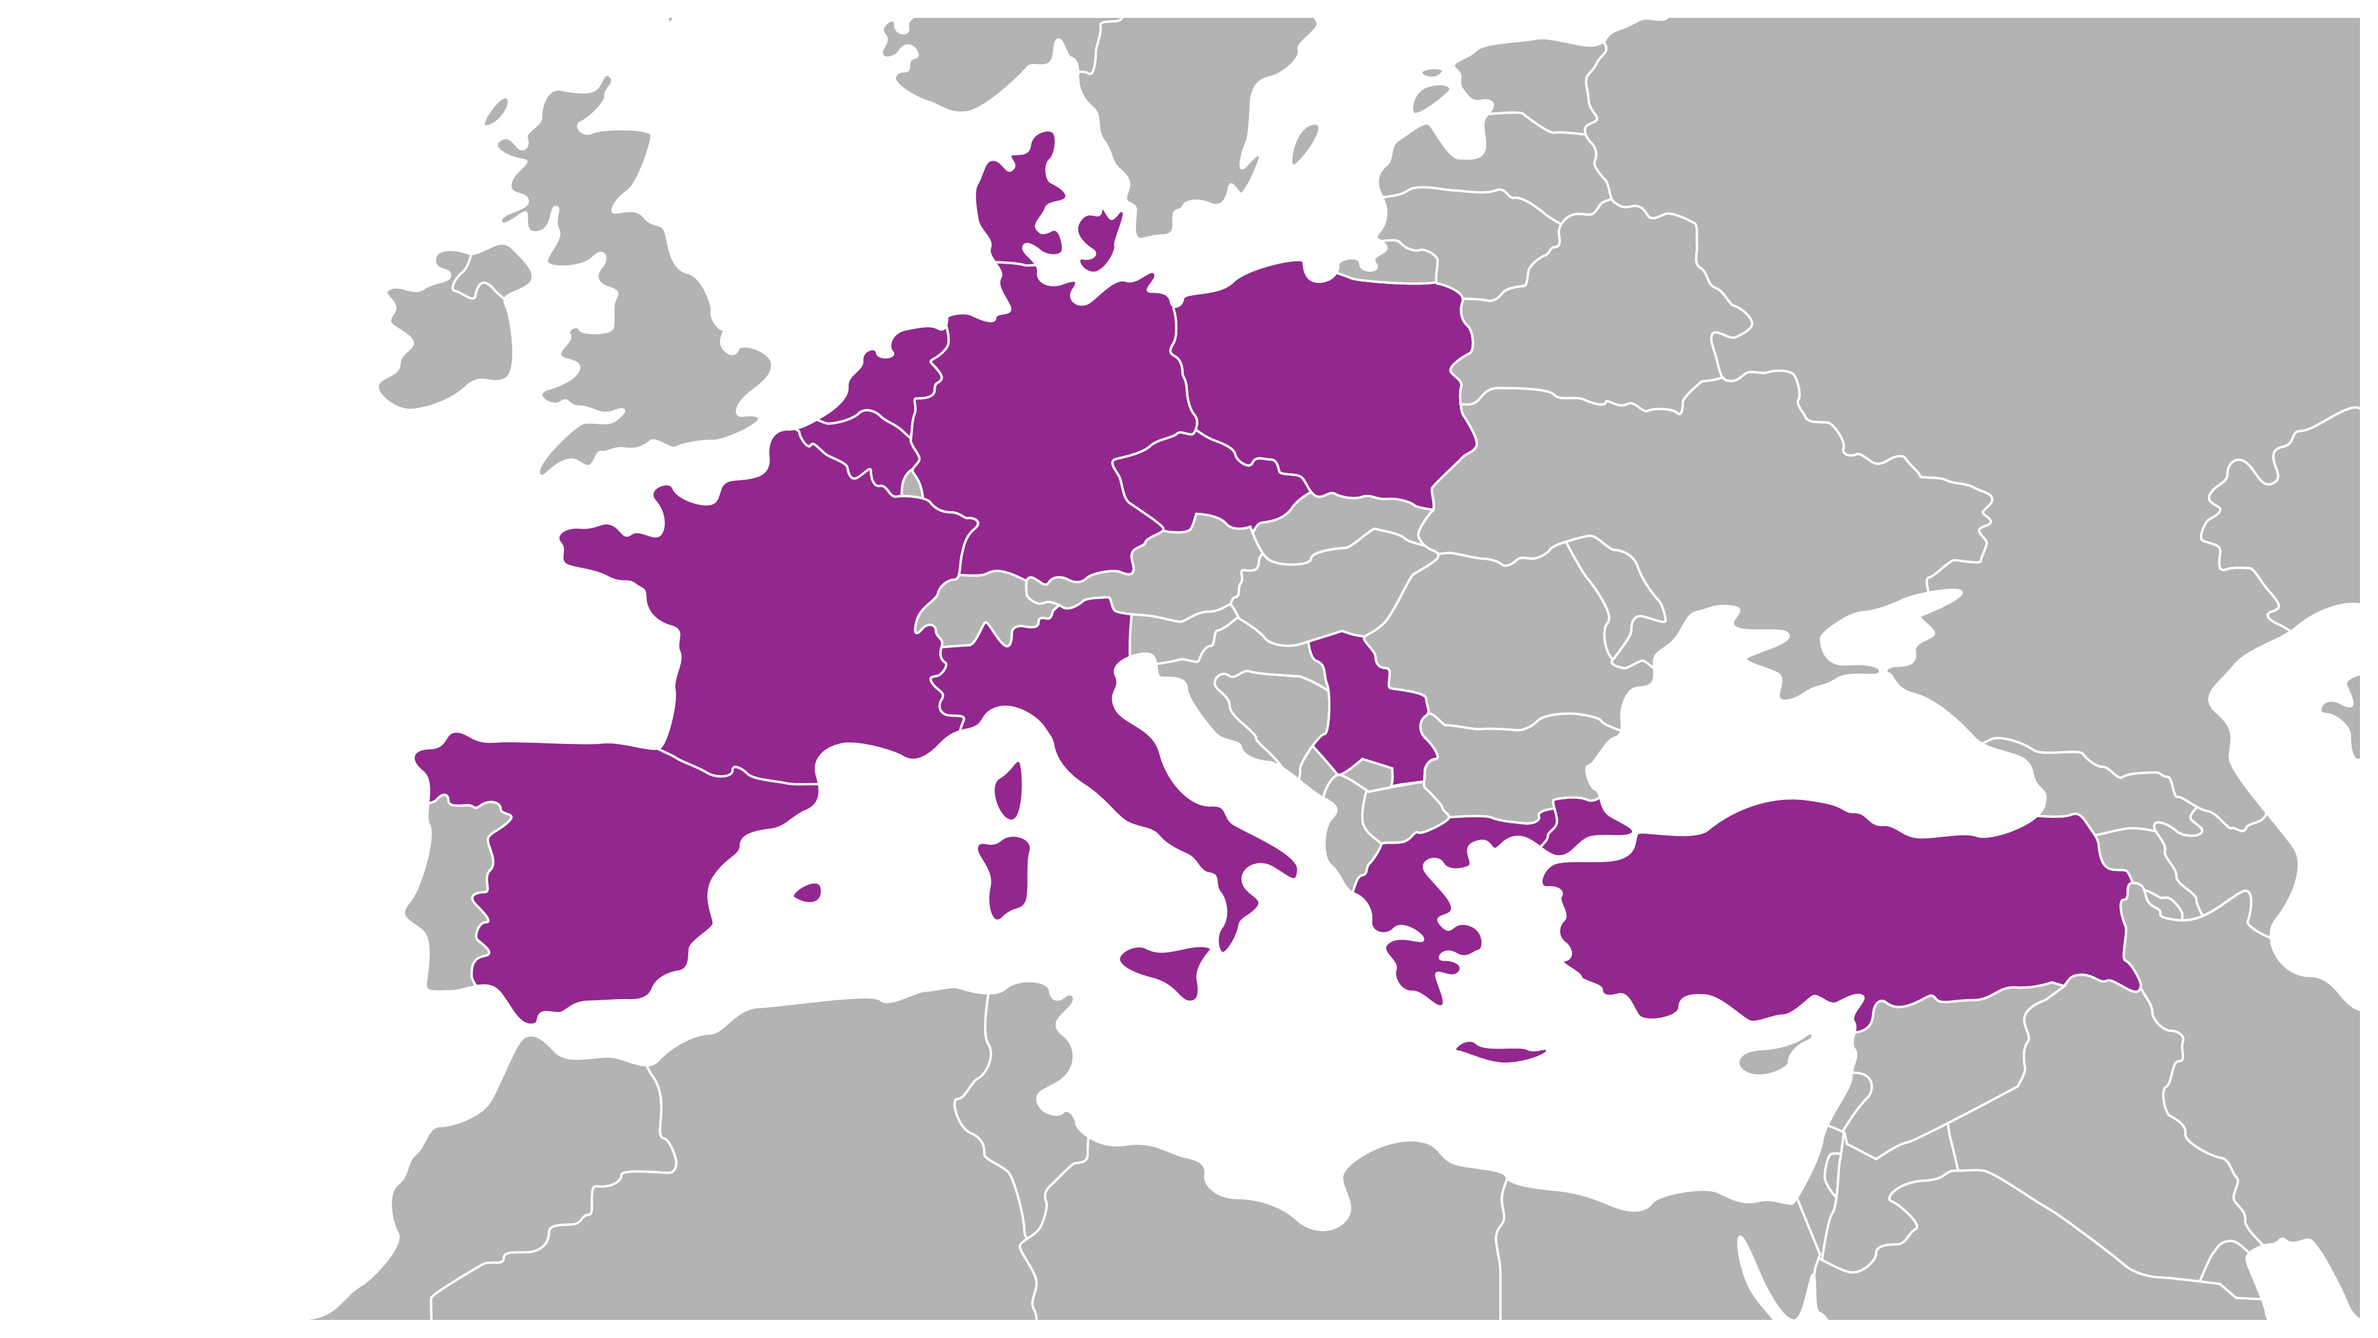

Study sites are in Belgium, Czech Republic, Denmark, France, Germany, Greece, Italy, The Netherlands, Poland, Serbia, Spain, and Turkey (shaded in purple).

Figure 4. APOLLO clinical sites.

Study Endpoints and Evaluations

- Primary
 - PFS
 - The primary analysis of PFS will occur after 188 PFS events are observed
- Secondary
 - Overall response rate and rates of ≥very good partial response and ≥complete response
 - Minimal residual disease negativity rate
 - Time to response
 - Duration of response
 - Time to next therapy
 - Overall survival
 - Safety
 - European Organization for Research and Treatment of Cancer Quality of Life Questionnaire (EORTC QLQ) Core 30 and EORTC QLQ Multiple Myeloma Module 20 scale and domain scores
 - European Quality of Life 5 Dimensions Questionnaire health utility values
 - Immunomodulatory effects of DARA on T cells
 - DARA serum concentration and immunogenicity

CONCLUSIONS

- **APOLLO is a phase 3, randomized, open-label, multicenter trial evaluating the efficacy and safety of Pom-Dex in combination with DARA SC in patients with RRMM**
- **This study is currently enrolling patients**

REFERENCES

1. de Weers M, et al. *J Immunol*. 2011;186(3):1840-1848.
2. Overdijk MB, et al. *MAbs*. 2015;7(2):311-321.
3. Overdijk MB, et al. *J Immunol*. 2016;197(3):807-813.
4. Lammerts van Bueren J, et al. *Blood*. 2014;124(21):3474.
5. Krejcik J, et al. *Blood*. 2016;128(3):384-394.
6. Quach H, et al. *Leukemia*. 2010;24(1):22-32.
7. POMALYST® (pomalidomide) capsules, for oral use [package insert]. Summit, NJ: Celgene Corporation; 2016.
8. Chari A, et al. *Blood*. 2017;130(8):974-981.
9. DARZALEX® (daratumumab) injection, for intravenous use [package insert]. Horsham, PA: Janssen Biotech, Inc; 2018.
10. Chari A, et al. Presented at: Annual Meeting of the American Society of Hematology (ASH); December 9-12, 2017; Atlanta, GA. Abstract 838.

ACKNOWLEDGMENTS

The authors would like to acknowledge the patients participating in this study and their families, as well as the global network of investigators, research nurses, study coordinators, and operations staff. This collaborative study (ClinicalTrials.gov Identifier: NCT03180736) is sponsored by the European Myeloma Network. Editorial and medical writing support were provided by Jason Jung, PhD, of MedErgy, and were funded by Janssen Global Services, LLC.

DISCLOSURES

PS consulted for, received honoraria from, and received research funding from Amgen, Celgene, Janssen, Karyopharm, and Takeda. ET received honoraria from Celgene, Novartis, Bristol-Myers Squibb, Janssen, Takeda, Amgen, and AbbVie; consulted for Amgen, Takeda, and Celgene; received research funding from Celgene, Amgen, and Janssen; and received travel expenses from Janssen, Amgen, Takeda, and Genesis. MAD received honoraria from and consulted for Celgene, Janssen, Takeda, and Amgen. JU, ES, NH, and JMS are employees of Janssen. JMS holds stock and/or stock options in J&J. EK received honoraria from Janssen, Celgene, Genesis, Takeda, Millennium, and Pharmacyclics; consulted for Janssen, Amgen, Takeda, and Prothena; and received research funding from Janssen and Amgen.

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ASCO® and the authors of this poster.
http://jjd_asco.scientificpresentations.org/Sonneveld_JJD63627.pdf

POSTER PRESENTED AT THE ANNUAL MEETING OF THE AMERICAN SOCIETY OF CLINICAL ONCOLOGY (ASCO); JUNE 1-5, 2018; CHICAGO, ILLINOIS.