1148P

A Phase I/II Safety Study of Tisotumab Vedotin (HuMax®-TF-ADC) in Patients With Solid Tumours

M. Chenard-Poirier[1]; D.S. Hong[2]; R.L. Coleman[3]; J. de Bono[1]; M. Mau-Sorensen[4]; D. Collins[1]; S. Lisby[5]*; L. Basse[5]*; U. Lassen[6]

[1]The Institute of Cancer Research/Royal Marsden NHS Foundation Trust, London, UK; [2]Phase I Unit, The University of Texas MD Anderson Cancer Center, Houston, TX, US; [3]Oncology, MD Anderson Cancer Center, Houston, TX, US; [4]Phase I Unit, Copenhagen University Hospital, Copenhagen, DK; [5]Medical Department, Genmab, Copenhagen, DK; [6]Department of Oncology, Phase 1 Unit, Rigshospitalet, Copenhagen, DK

*Dr Lisby and Dr Basse were Genmab employees when the research was conducted and during the writing of the abstract.

BACKGROUND[1,2]

- Tissue factor (TF; CD142; thromboplastin) is
 - A transmembrane protein that is the main physiological initiator of coagulation and is involved in angiogenesis, cell adhesion, motility, and cell survival
 - Sequestered from the circulation under normal conditions
 - Aberrantly expressed and associated with poor prognosis in solid cancers
 - An attractive antibody-drug conjugate (ADC) target due to optimal internalisation characteristics
- Tisotumab vedotin is an ADC composed of
 - A human monoclonal antibody (mAb) specific for TF
 - The microtubule disrupting agent monomethyl auristatin E (MMAE)[a]
 - A protease-cleavable linker that covalently attaches MMAE to TF mAb
- Tisotumab vedotin selectively targets TF to deliver a clinically validated toxic payload to tumour cells **(Figure 1)**

Figure 1: Tisotumab Vedotin Mechanism of Action[1,3]

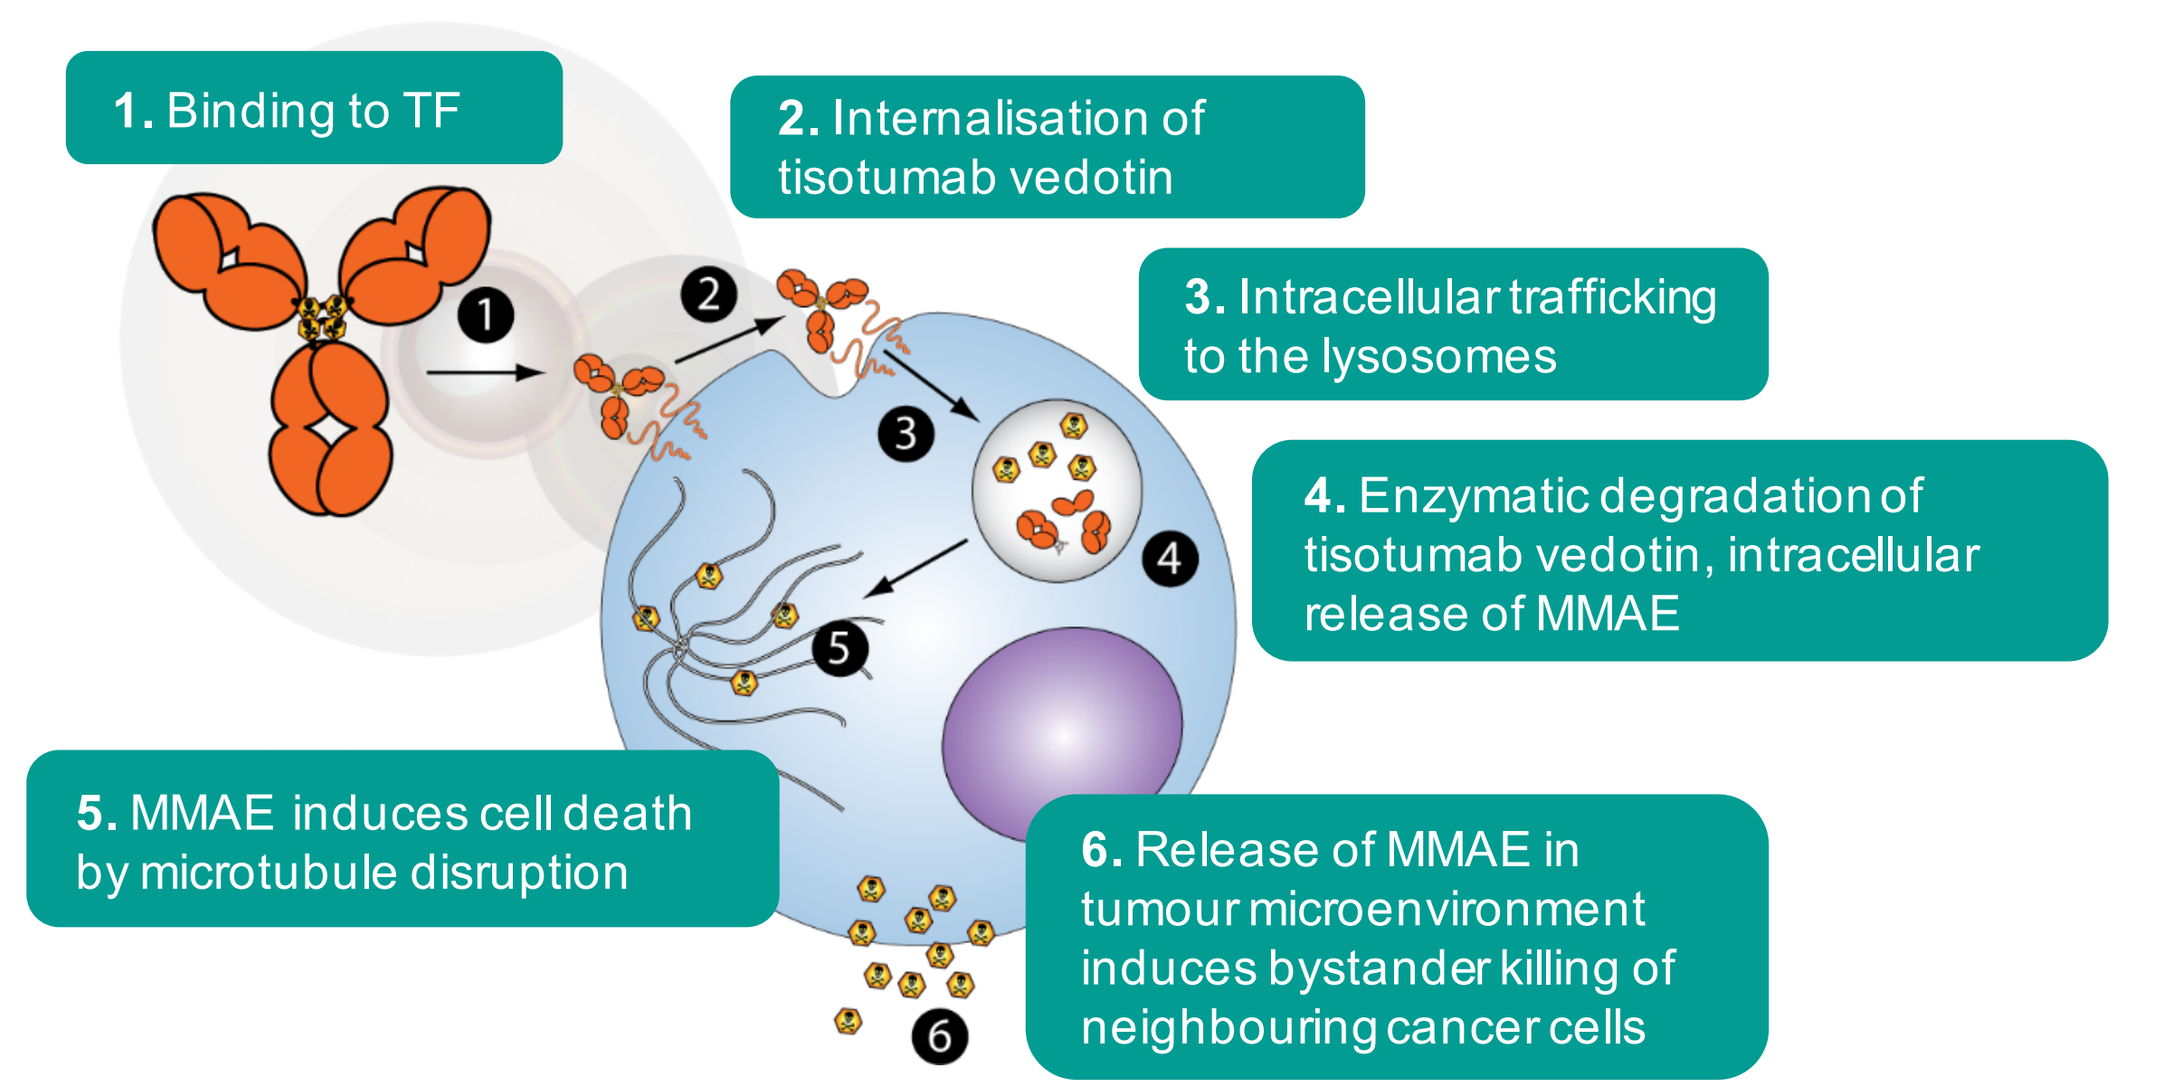
1. Binding to TF
2. Internalisation of tisotumab vedotin
3. Intracellular trafficking to the lysosomes
4. Enzymatic degradation of tisotumab vedotin, intracellular release of MMAE
5. MMAE induces cell death by microtubule disruption
6. Release of MMAE in tumour microenvironment induces bystander killing of neighbouring cancer cells

- Tisotumab vedotin showed potent anti-tumour activity in patient-derived xenograft models for a broad range of solid cancers, including cervical cancer

[a]MMAE-based ADC technology was licensed from Seattle Genetics, Inc., in a license and collaboration agreement.

METHODS

GEN701 Study Overview[4]

- First-in-human phase I/II dose-escalating and expansion safety study of tisotumab vedotin in patients with locally advanced and/or metastatic solid tumours known to express TF **(Figure 2)**
- Open-label, multicentre, single-arm study (NCT02001623)
- Safety and efficacy data are presented for only the dose escalation portion of the study

Figure 2: GEN701 Part 1 and Part 2 Study Design Overview

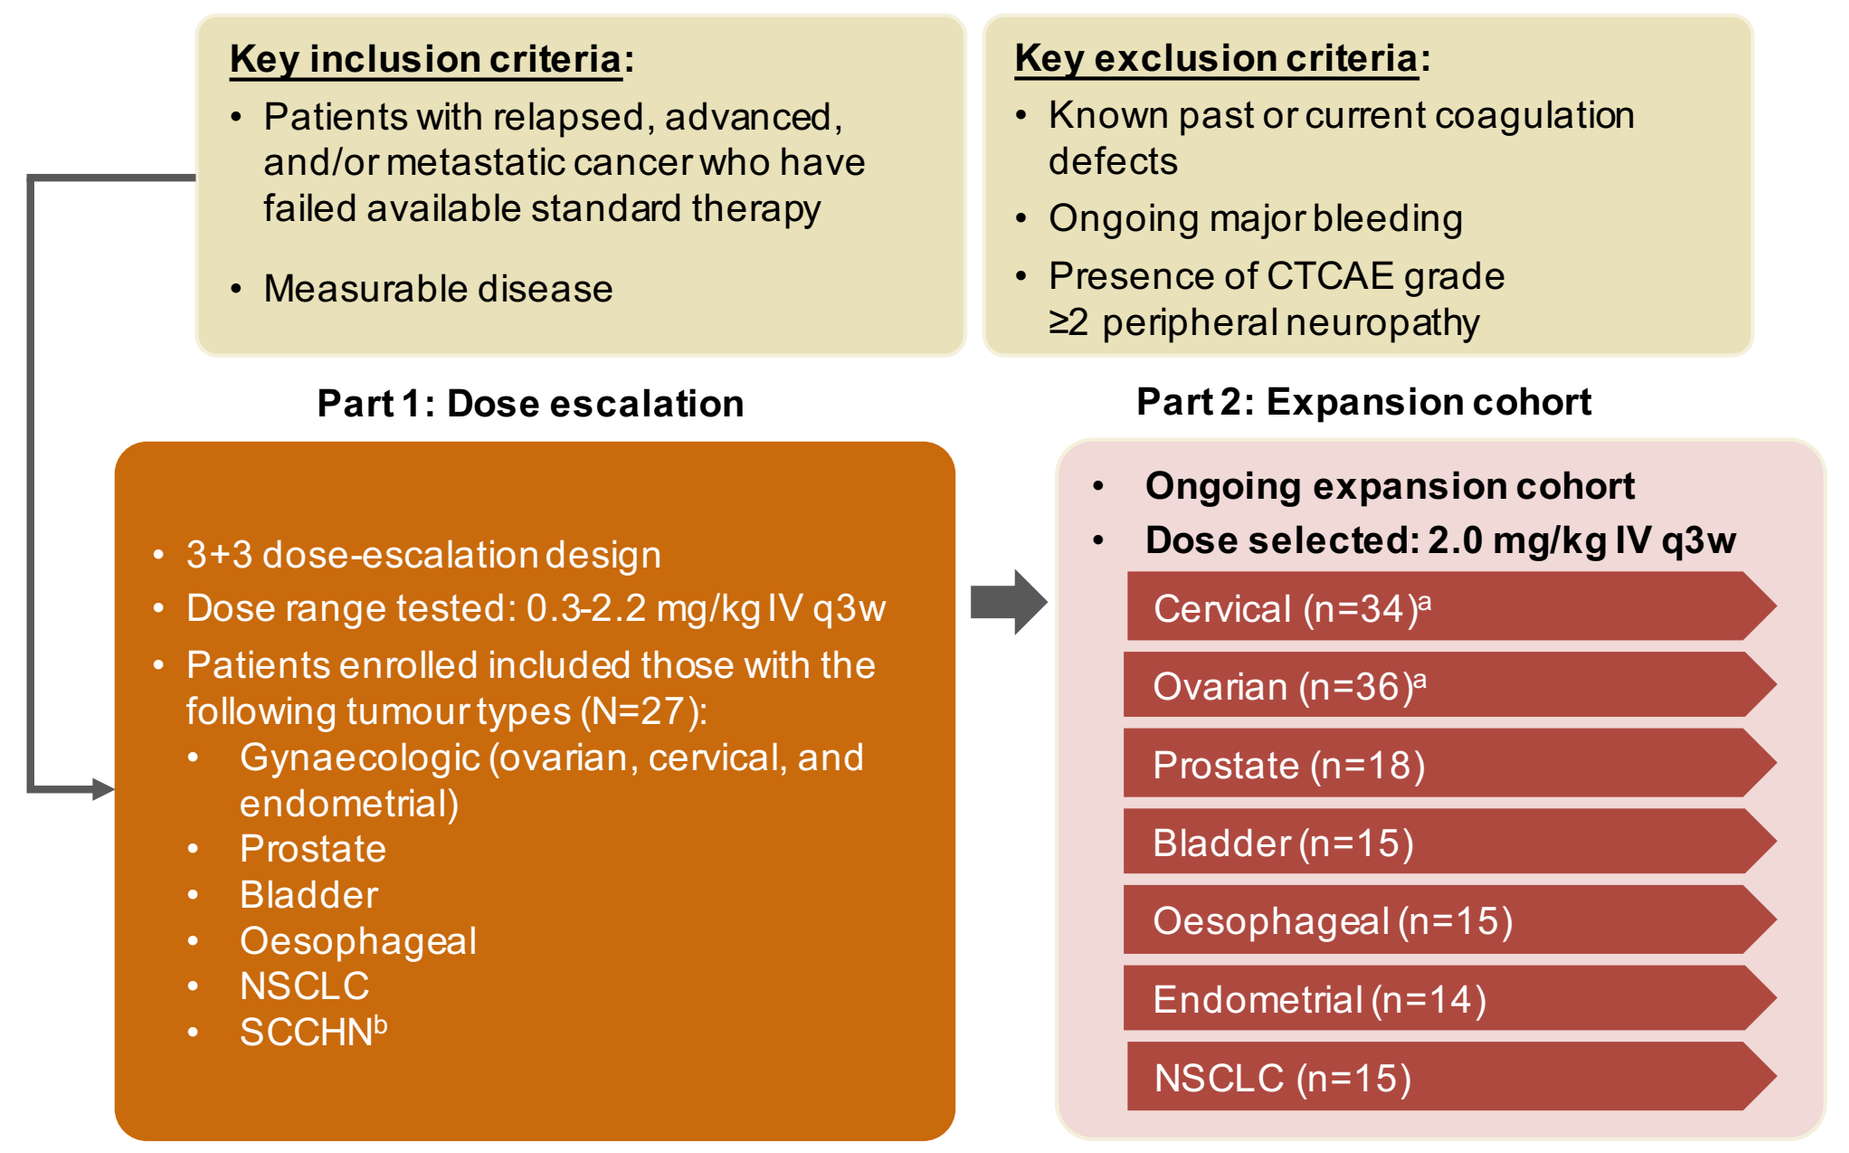
Key inclusion criteria:
• Patients with relapsed, advanced, and/or metastatic cancer who have failed available standard therapy
• Measurable disease
Key exclusion criteria:
• Known past or current coagulation defects
• Ongoing major bleeding
• Presence of CTCAE grade ≥2 peripheral neuropathy
Part 1: Dose escalation
• 3+3 dose-escalation design
• Dose range tested: 0.3-2.2 mg/kg IV q3w
• Patients enrolled included those with the following tumour types (N=27):
• Gynaecologic (ovarian, cervical, and endometrial)
• Prostate
• Bladder
• Oesophageal
• NSCLC
• SCCHN[b]
Part 2: Expansion cohort
• Ongoing expansion cohort
• Dose selected: 2.0 mg/kg IV q3w
Cervical (n=34)[a]
Ovarian (n=36)[a]
Prostate (n=18)
Bladder (n=15)
Oesophageal (n=15)
Endometrial (n=14)
NSCLC (n=15)

[a]In phase II, ovarian and cervical cohorts were expanded to approximately 30 patients based on preliminary efficacy observed in the first 14 patients enrolled. [b]The SCCHN cohort was closed by protocol amendment 4 due to an event of pharyngeal tumour haemorrhage with fatal outcome. The event was deemed to be most likely related to the disease itself.

CTCAE=Common Terminology Criteria for Adverse Events; IV=intravenous; q3w=every 3 weeks; NSCLC=non–small cell lung cancer; SCCHN=squamous cell carcinoma of the head and neck.

GEN701 Part 1, Dose Escalation: Study Design[4]

- GEN701 part 1 is a dose-escalation study following a 3+3 design **(Figure 3)**

Figure 3: GEN701 Part 1 Study Design Overview

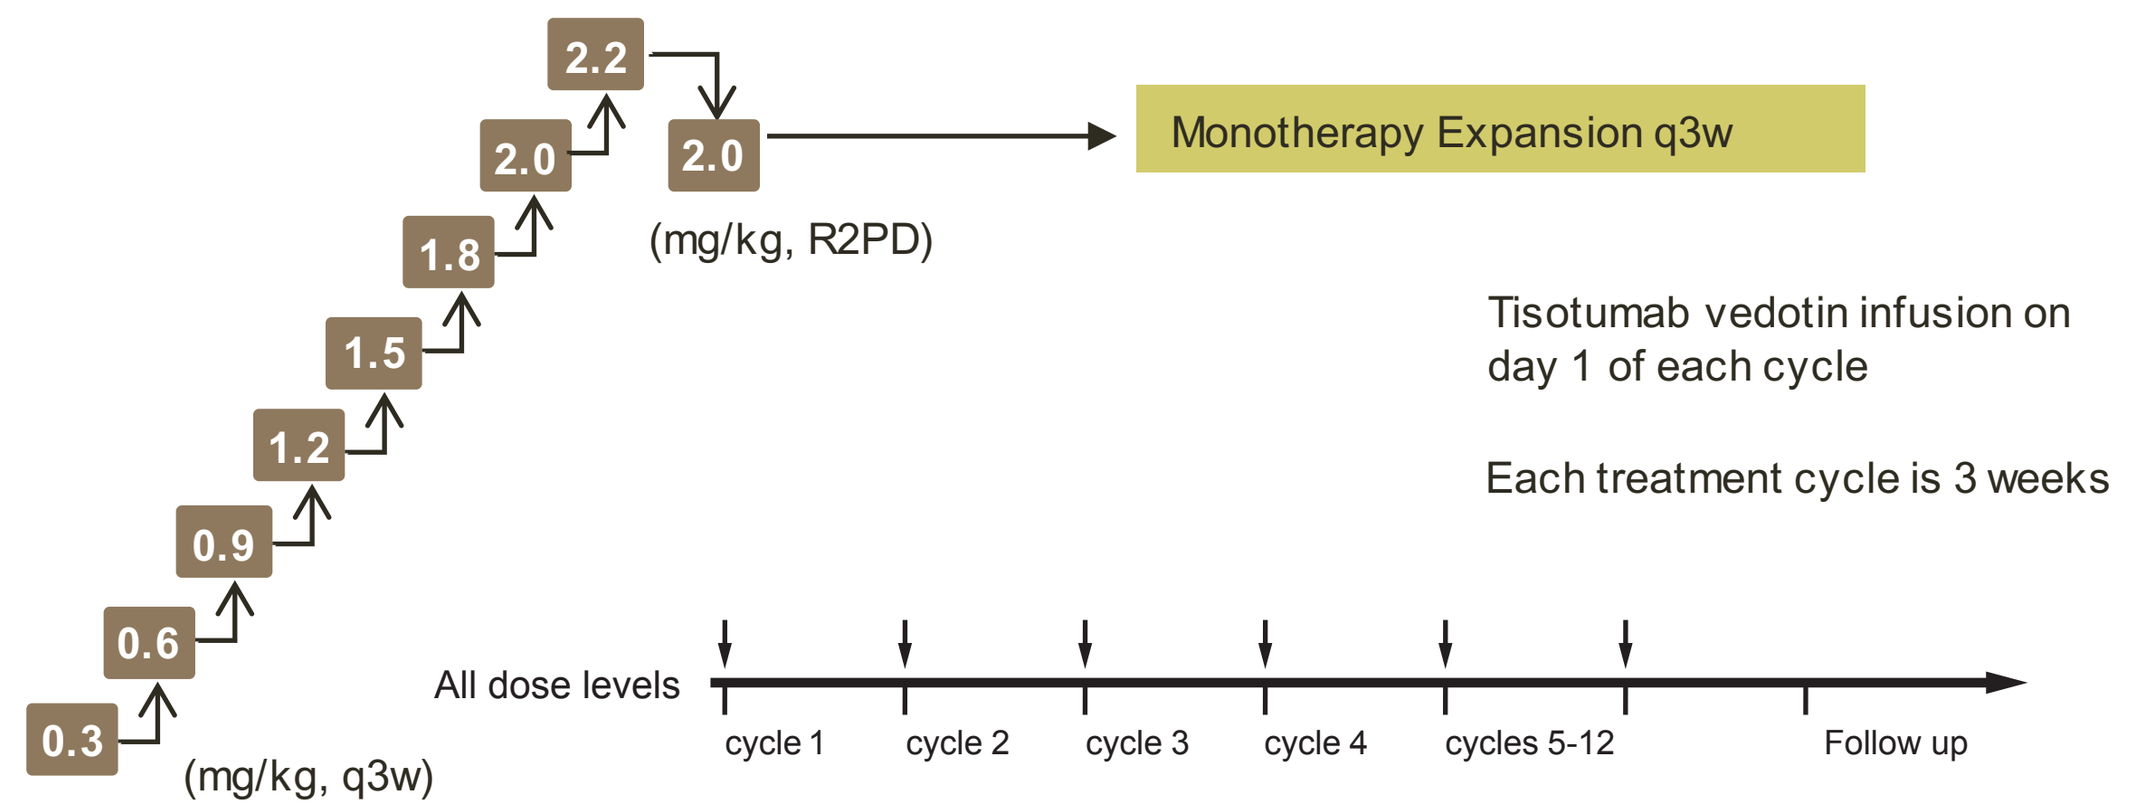
0.3
0.6
0.9
1.2
1.5
1.8
2.0
2.2
(mg/kg, q3w)
2.0
(mg/kg, R2PD)
Monotherapy Expansion q3w
Tisotumab vedotin infusion on day 1 of each cycle
Each treatment cycle is 3 weeks
All dose levels
cycle 1
cycle 2
cycle 3
cycle 4
cycles 5-12
Follow up

- Tisotumab vedotin was administered by IV infusion at doses ranging from 0.3 to 2.2 mg/kg on day 1 of a 21-day cycle for 4 cycles
 - Patients with SD or better at the end of 4 cycles had the option to continue tisotumab vedotin for 8 additional cycles
- Dose-limiting toxicities (DLTs) were determined during the first cycle and were defined as grade ≥3 events possibly related to study drug
- Maximum tolerated dose (MTD) was defined as the highest tisotumab vedotin dose level that does not cause unacceptable side effects
- The recommended phase II dose (RP2D) of tisotumab vedotin was further evaluated for safety and anti-tumour activity in several expansion phase cohorts as per the trial design schema
- Tumor biopsies were required at baseline for TF expression[a]

[a]TF expression was assessed by immunohistochemistry utilising the TF mAb antibody. TF staining intensity was determined using the H-scoring system.

Objectives

Primary objective

- Assess the safety and tolerability in a mixed population of patients with specified solid tumours (Adverse event [AE] severity was graded according to Common Terminology Criteria for Adverse Events [CTCAE] version 4.03)

Secondary objectives

- Determine the pharmacokinetic (PK) profile
- Establish the MTD and the RP2D
- Evaluate preliminary anti-tumour activity (assessed according to Response Evaluation Criteria in Solid Tumors [RECIST] version 1.1; tumour evaluations were performed by CT scans every 6 weeks)[a]

[a]In order to qualify for SD, results of the CT scan scheduled for week 6 needed to be SD or better. Two CT scans were performed outside the per-protocol defined window.

RESULTS

Patient Population

- Patient demographics and baseline characteristics are shown in **Table 1**
- A total of 25 patients withdrew from treatment due to patient choice (4%), disease progression (67%), DLT (4%), AEs (15%), or death (4%); 2 patients continued therapy beyond 4 cycles

Table 1: Patient Demographics and Baseline Characteristics

Characteristic		All Patients (N=27)
Age, median (range), years		62 (43-73)
Gender, n (%)	Male	9 (33)
	Female	18 (67)
ECOG PS, n (%)	0	13 (48)
	1	13 (48)
	NA	1 (4)
Primary tumour type, n (%)	Ovary	7 (26)
	Cervix	2 (7)
	Endometrium	3 (11)
	Bladder	2 (7)
	Prostate	4 (15)
	Oesophagus	4 (15)
	Head and neck	1 (4)
	Lung	4 (15)
Median number of prior therapies (range)		3 (1-14)

ECOG PS=Eastern Cooperative Oncology Group; IHC=immunohistochemistry; PS=performance status; TF=tissue factor.

Safety

- Safety analyses are available for all 27 patients and are reported in **Table 2**
- 25 patients (93%) experienced treatment-related AEs, the most common of which were fatigue (48%), epistaxis (48%), and anaemia (41%) **(Table 2; Figure 4)**
- 19 patients (70%) experienced grade ≥3 treatment-related AEs, the most common of which were fatigue (n=4), anaemia (n=4), abdominal pain (n=3), hyponatraemia (n=3) **(Table 2)**
 - There were no grade 4 events
- 7 patients discontinued due to AEs, which included grade 1 pneumonitis (n=1), grade 3 events for Guillain-Barré syndrome (n=1), diabetes mellitus (n=1), fatigue (n=1), and abdominal pain (n=2), and 1 patient experienced grade 2 peripheral swelling and grade 3 pain in extremity **(Table 2)**
- There were 3 deaths reported in this study **(Table 2)**
 - 1 patient with squamous cell cancer of the head and neck in the 0.6 mg/kg cohort died from tumour-related bleeding
 - 2 patients in the 0.3 mg/kg cohort died from disease progression, both were considered not related to the study drug
- No significant changes in coagulation parameters have been observed
 - The mean prothrombin time at baseline was 11.5 seconds (n=18) and 11.7 seconds (n=17) by the end of the study
 - The mean activated partial thromboplastin time at baseline was 28.2 seconds (n=25) and 27.1 seconds (n=23) by the end of the study

Table 2: Overall Safety Profile of Tisotumab Vedotin by Dose Cohorts

AE Category, n (%)[a]	All Doses (N=27)	0.3 mg/kg (n=3)	0.6 mg/kg (n=3)	0.9 mg/kg (n=3)	1.2 mg/kg (n=3)	1.5 mg/kg (n=3)	1.8 mg/kg (n=3)	2.0 mg/kg (n=3)	2.2 mg/kg (n=6)
AE	27 (100)	3 (100)	3 (100)	3 (100)	3 (100)	3 (100)	3 (100)	3 (100)	6 (100)
Serious AE	15 (56)	2 (67)	1 (33)	0	2 (67)	2 (67)	2 (67)	2 (67)	4 (67)
Grade ≥3 AE	19 (70)	2 (67)	3 (100)	2 (67)	1 (33)	2 (67)	3 (100)	2 (67)	4 (67)
Treatment-related AE	25 (93)	3 (100)	3 (100)	1 (33)	3 (100)	3 (100)	3 (100)	3 (100)	6 (100)
AE leading to discontinuation	7 (26)	0	0	0	0	0	2 (67)	0	5 (83)
AE with outcome of death	3 (11)	2 (67)	1 (33)	0	0	0	0	0	0

[a]Occurring up to 30 days after the last treatment.
AE=adverse event.

Figure 4: Most Common Treatment-Related AEs (Occurring in ≥4 Patients Overall) With Tisotumab Vedotin (All Doses)

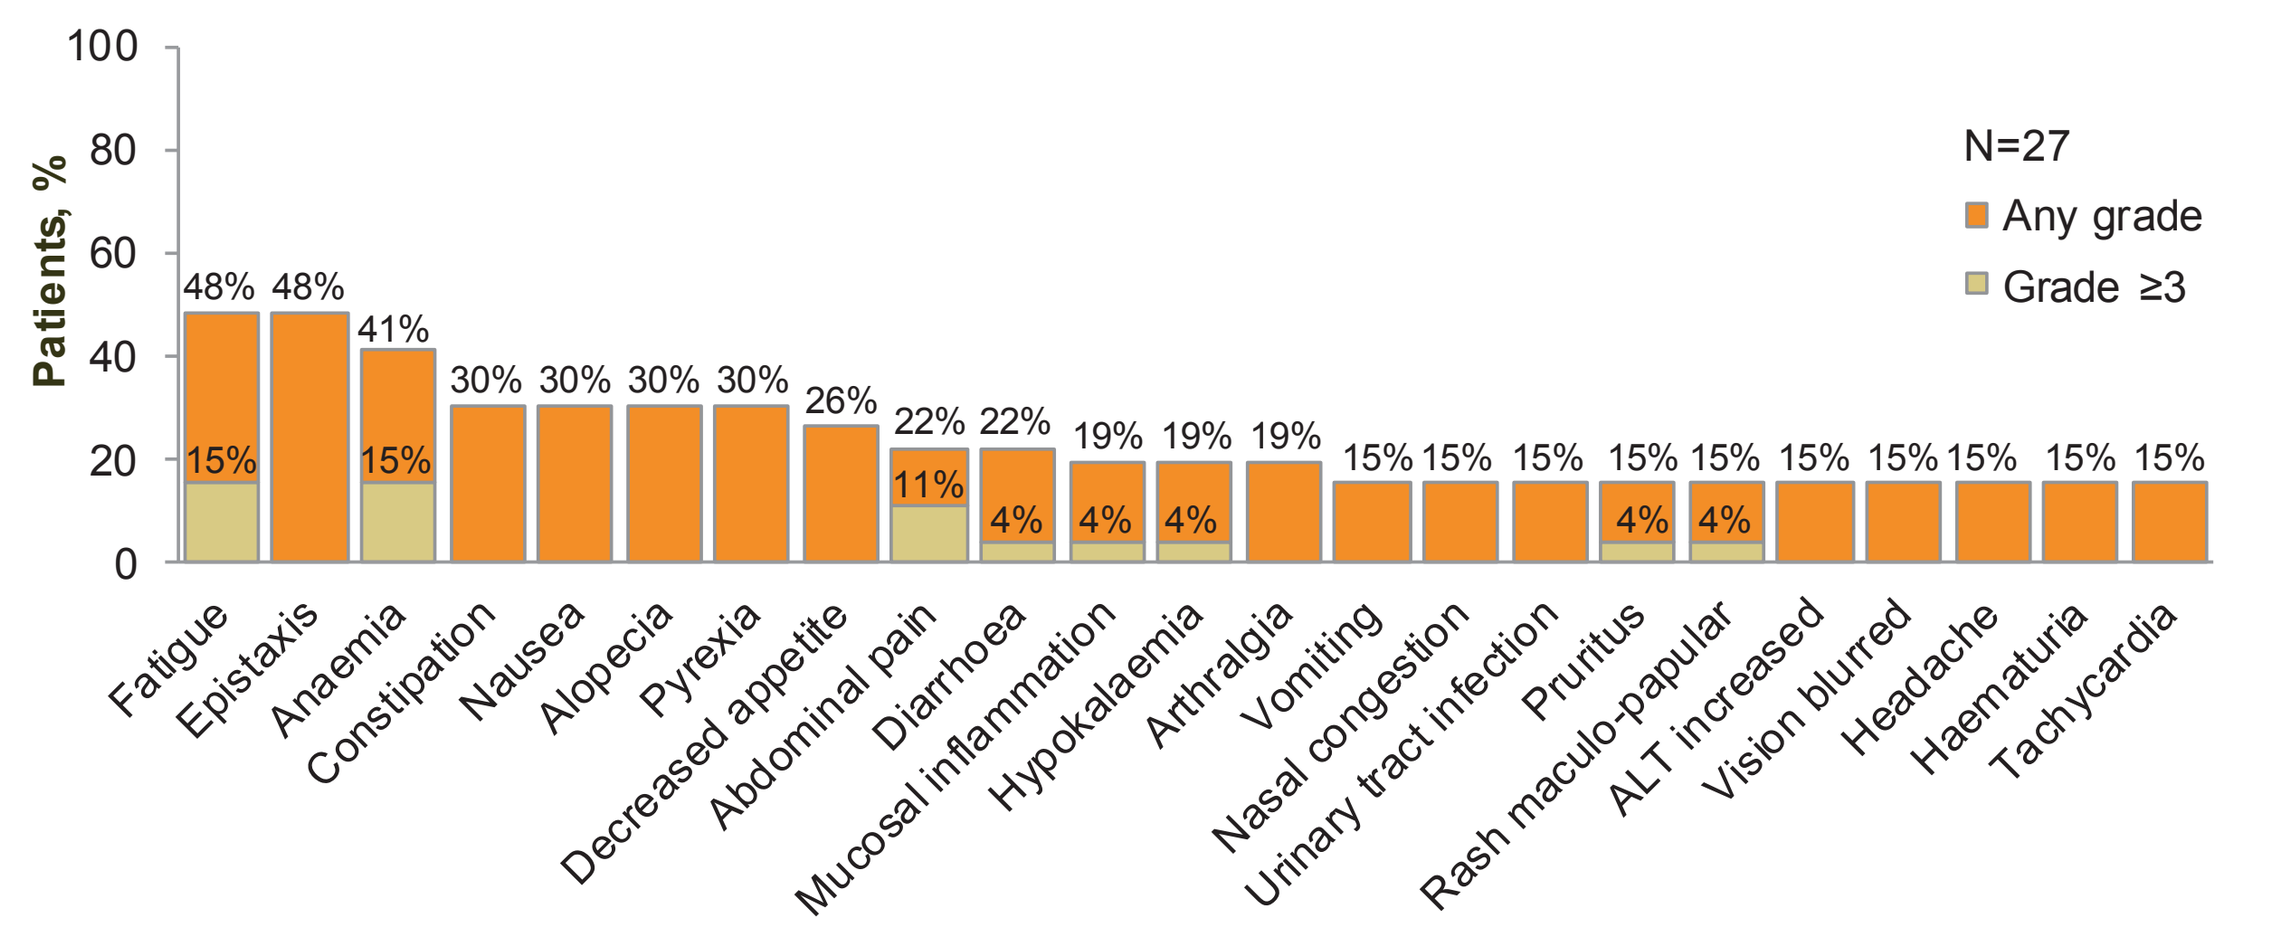
Patients, %
100
80
60
40
20
0
N=27
Any grade
Grade ≥3
48% 48% 41% 30% 30% 30% 30% 26% 22% 22% 19% 19% 19% 15% 15% 15% 15% 15% 15% 15% 15% 15% 15%
15% 15% 11% 4% 4% 4% 4% 4%
Fatigue
Epistaxis
Anaemia
Constipation
Nausea
Alopecia
Pyrexia
Decreased appetite
Abdominal pain
Diarrhoea
Mucosal inflammation
Hypokalaemia
Arthralgia
Vomiting
Nasal congestion
Urinary tract infection
Pruritus
Rash maculo-papular
ALT increased
Vision blurred
Headache
Haematuria
Tachycardia

MTD and RP2D Determination

- Three DLTs (diabetes mellitus type 2, mucositis, and neutropenic fever, all grade 3) were seen in 3 patients in the 2.2 mg/kg dose cohort
- The MTD was identified as 2.0 mg/kg
- The dose for the expansion phase was defined as 2.0 mg/kg

Pharmacokinetics

- After a single dose of tisotumab vedotin at 2.0 mg/kg, geometric means (CV%) of the time to reach C_{max} (T_{max}) (hr), maximum concentration (C_{max}) (ng/mL), and area under the concentration time curve $(AUC)_{0\text{-}t}$ (hr*ng/mL) were 1.2 (8%), 32296.1 (22%), and 1256379.7 (33%), respectively **(Table 3)**
- Low levels of unconjugated MMAE are measured in systemic circulation **(Figure 5)**

Table 3: Summary of Tisotumab Vedotin Plasma PK Parameters by Dose Cohorts in Cycle 1

Dose (mg/kg)	n	T_{max} (hr)	C_{max} (ng/mL)	$AUC_{0\text{-}t}$ (hr*ng/mL)
0.3	3	1.5 (73%)	4782.7 (12%)	59216.8 (3%)
0.6	3	1.2 (13%)	12195.3 (10%)	368432.7 (8%)
0.9	3	1.3 (12%)	19811.6 (17%)	601926.2 (17%)
1.2	3	1.3 (12%)	34673.1 (19%)	1084672.7 (9%)
1.5	3	1.1 (9.6%)	23115.6 (21%)	794988.4 (19%)
1.8	3	1.2 (14%)	35416.3 (39%)	1504823.8 (50%)
2.0	3	1.2 (8%)	32296.1 (22%)	1256379.7 (33%)
2.2	6	1.1 (13%)	55530.3 (10%)	2037070.5 (34%)

Values for the PK parameters are for geometric mean (CV%).
AUC=area under the curve; C_{max}=maximum concentration; T_{max}=time taken to reach C_{max}; PK=pharmacokinetic.

Figure 5: Mean Plasma Tisotumab Vedotin and Free MMAE Concentration-Time Profiles for Cycles 1 and 2

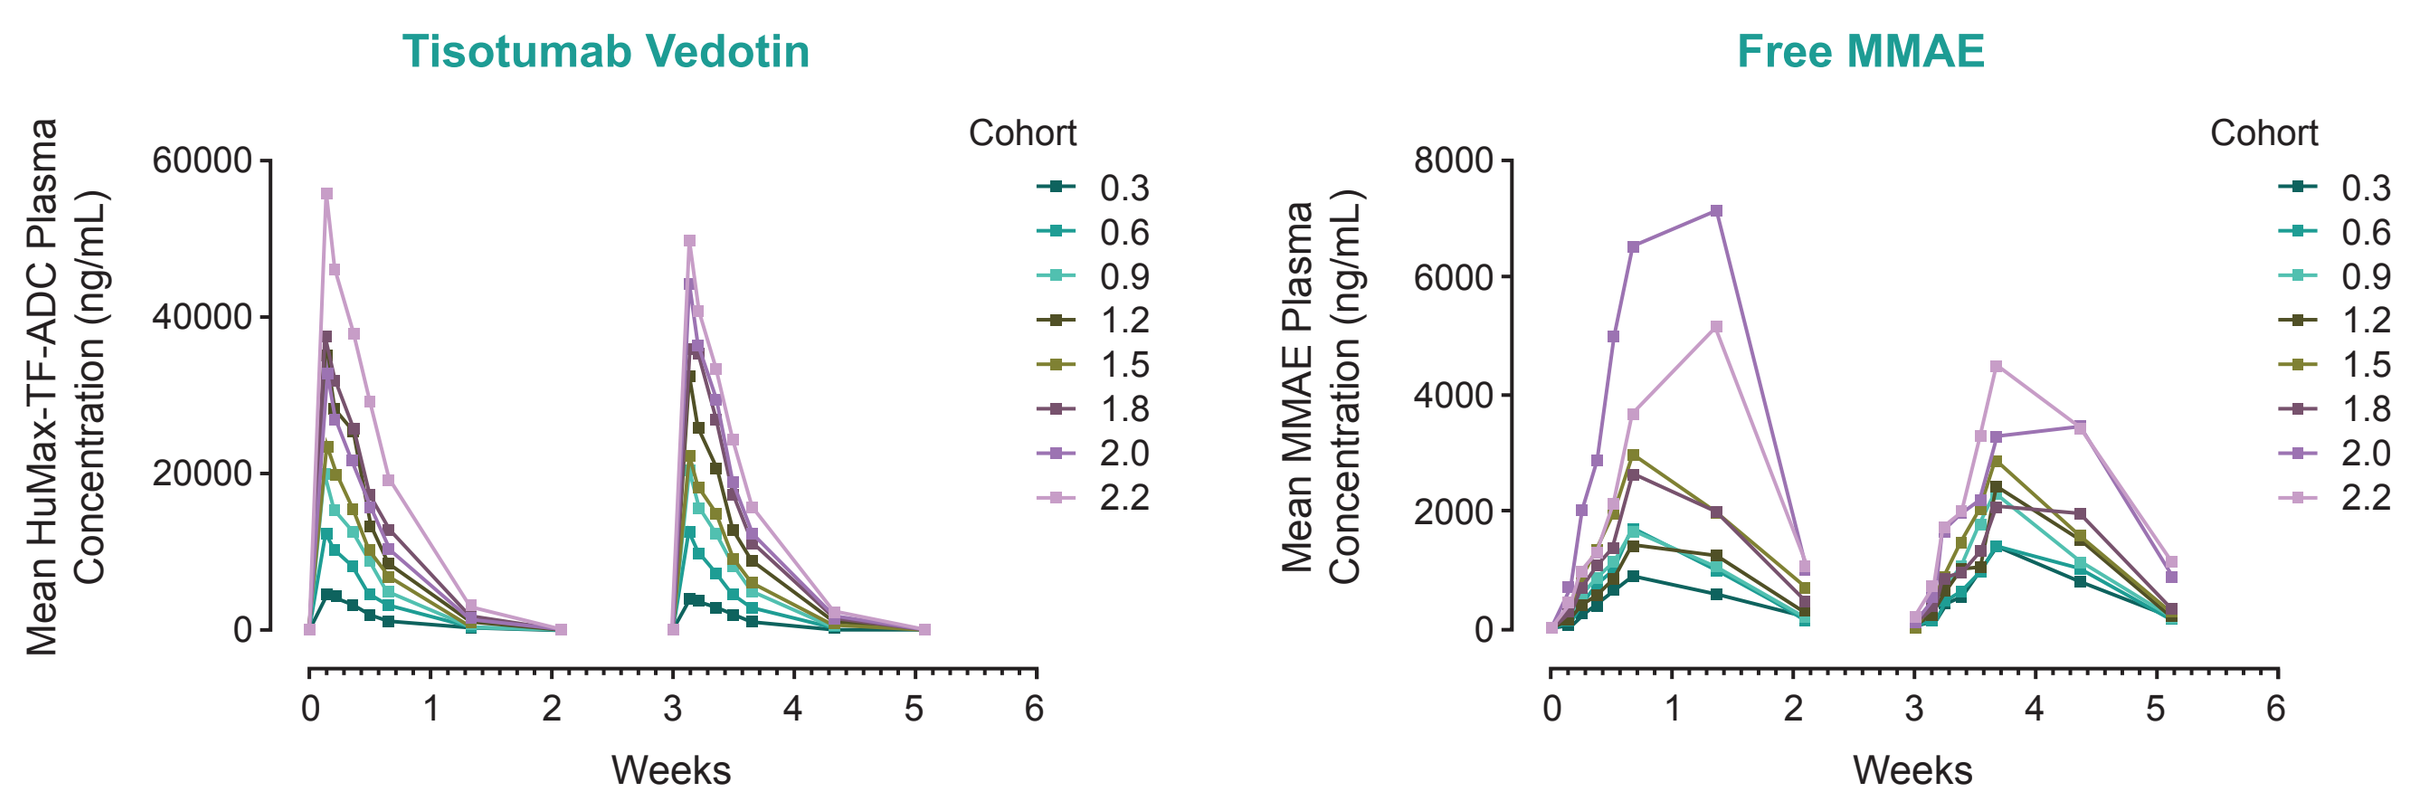
Tisotumab Vedotin
Mean HuMax-TF-ADC Plasma Concentration (ng/mL)
Free MMAE
Mean MMAE Plasma Concentration (ng/mL)
Weeks
Cohort
0.3
0.6
0.9
1.2
1.5
1.8
2.0
2.2

MMAE=monomethyl auristatin.

Efficacy

- 26 patients were evaluated for efficacy **(Table 4)**
- The best response observed was PR in 1 patient (4%) and SD in 11 patients (41%) **(Table 4)**
- Disease control rate (DCR; partial response [PR] + SD) was 46% (12/26)
- Changes in tumour size, expressed as a percentage of baseline, are shown in **Figure 6**

Table 4: Confirmed Objective Responses (Investigator-Assessed)

Confirmed Response per RECIST v1.1, n (%)[a]	All Doses (N=27)	0.3 mg/kg (n=3)	0.6 mg/kg (n=3)	0.9 mg/kg (n=3)	1.2 mg/kg (n=3)	1.5 mg/kg (n=3)	1.8 mg/kg (n=3)	2.0 mg/kg (n=3)	2.2 mg/kg (n=6)
Complete response	**0**	0	0	0	0	0	0	0	0
Partial response	**1 (4)**	0	0	0	1 (33)	0	0	0	0
Stable disease	**11 (41)**	0	1 (33)	1 (33)	1 (33)	0	3 (100)	1 (33)	4 (67)
Progressive disease	**14 (52)**	3 (100)	1 (33)	2 (67)	1 (33)	3 (100)	0	2 (67)	2 (33)
Not evaluable[b]	**1 (4)**	0	1 (33)	0	0	0	0	0	0

[a]Percentages may not add to 100% due to rounding. [b]Patient died prior to the first scan.

Figure 6: Best Percentage Change in Tumour Size From Baseline

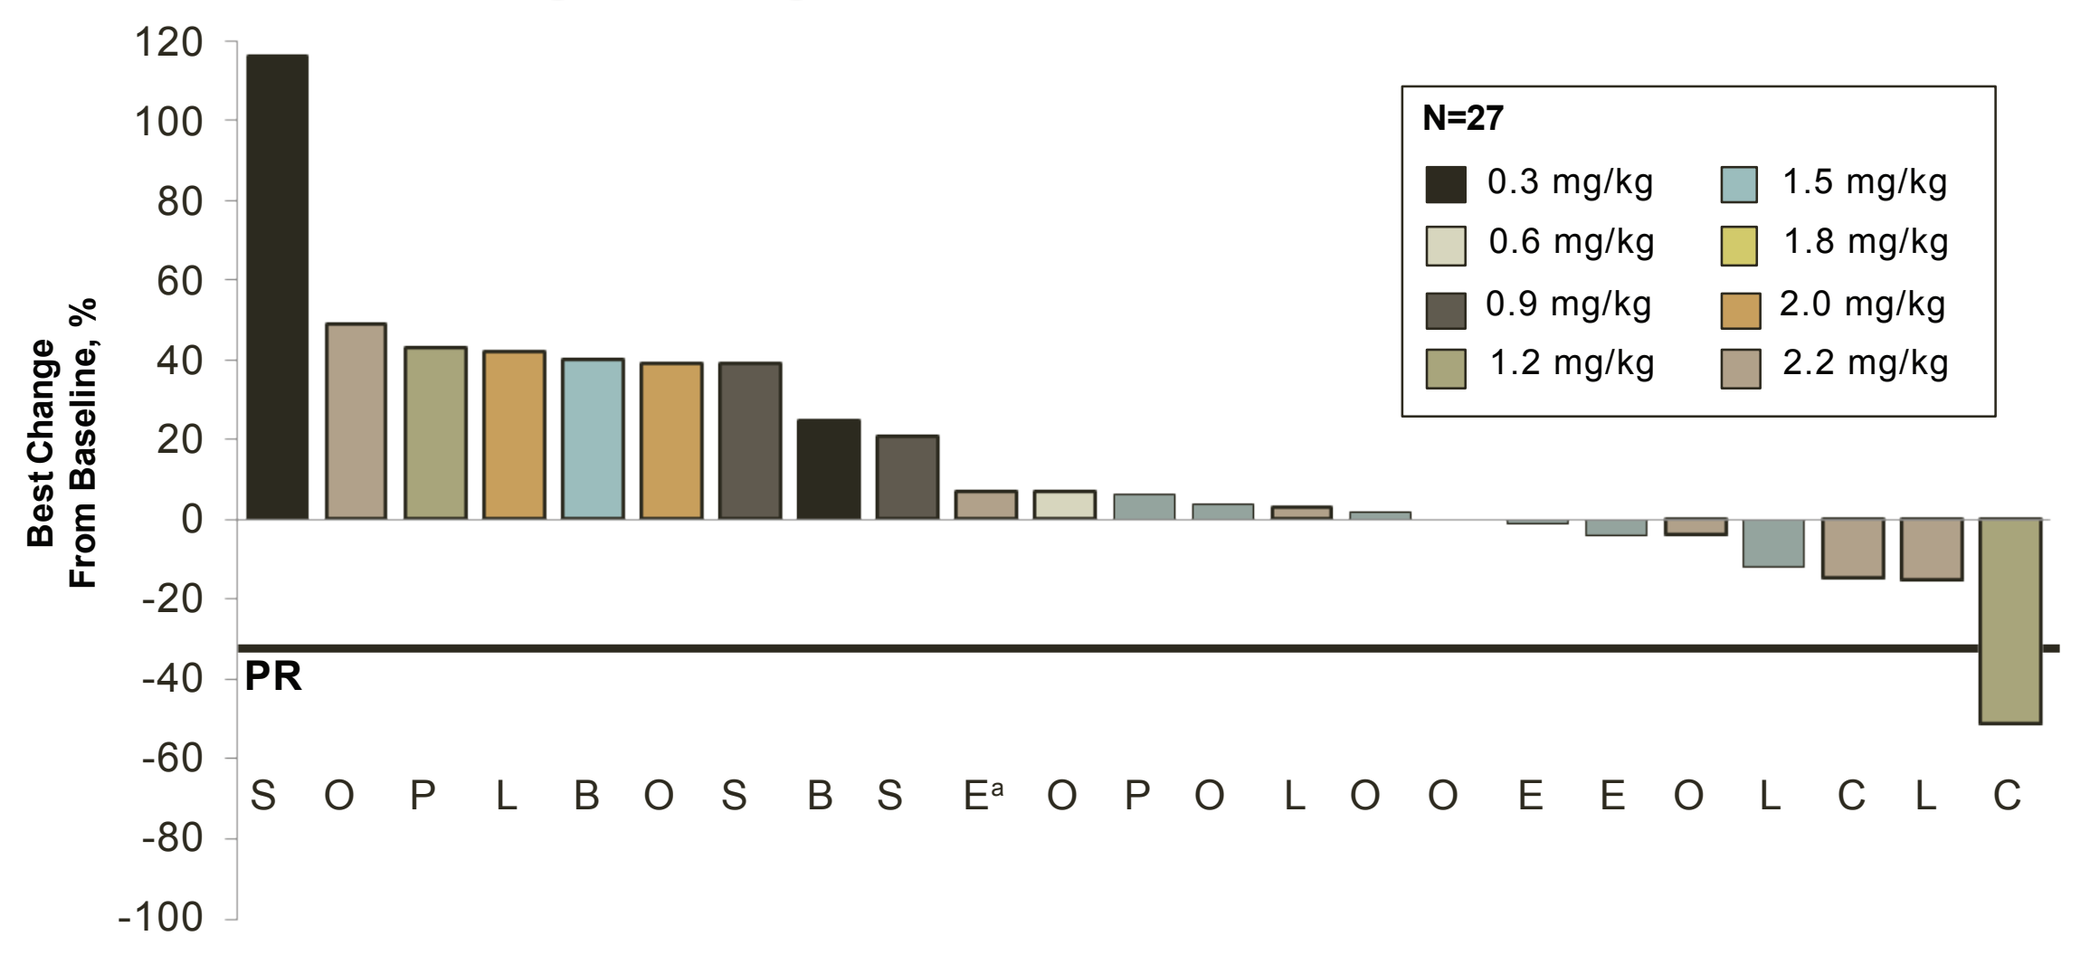
Best Change From Baseline, %
120
100
80
60
40
20
0
-20
-40
-60
-80
-100
PR
N=27
0.3 mg/kg
0.6 mg/kg
0.9 mg/kg
1.2 mg/kg
1.5 mg/kg
1.8 mg/kg
2.0 mg/kg
2.2 mg/kg
S O P L B O S B S E[a] O P O L O O E E O L C L C

[a]Patient experienced disease progression by virtue of a new lesion.
B=bladder; C=cervix; E=endometrium; L=lung; O=ovary; P=prostate; S=squamous cell carcinoma of the head and neck.

CASE STUDY

- 43-year-old cervical cancer patient diagnosed with stage 4 disease who had received 3 prior lines of therapy
- TF expression: H-score 140 (archival)
- Achieved a confirmed PR, with a 51% reduction in the target lesion and continued benefit for a total of 15 months **(Figure 7)**
- Tisotumab vedotin was well tolerated and no severe AEs were reported
- The patient eventually experienced disease progression and stopped therapy

Figure 7: CT Scan of Lung Metastasis of Patient Treated With Tisotumab Vedotin

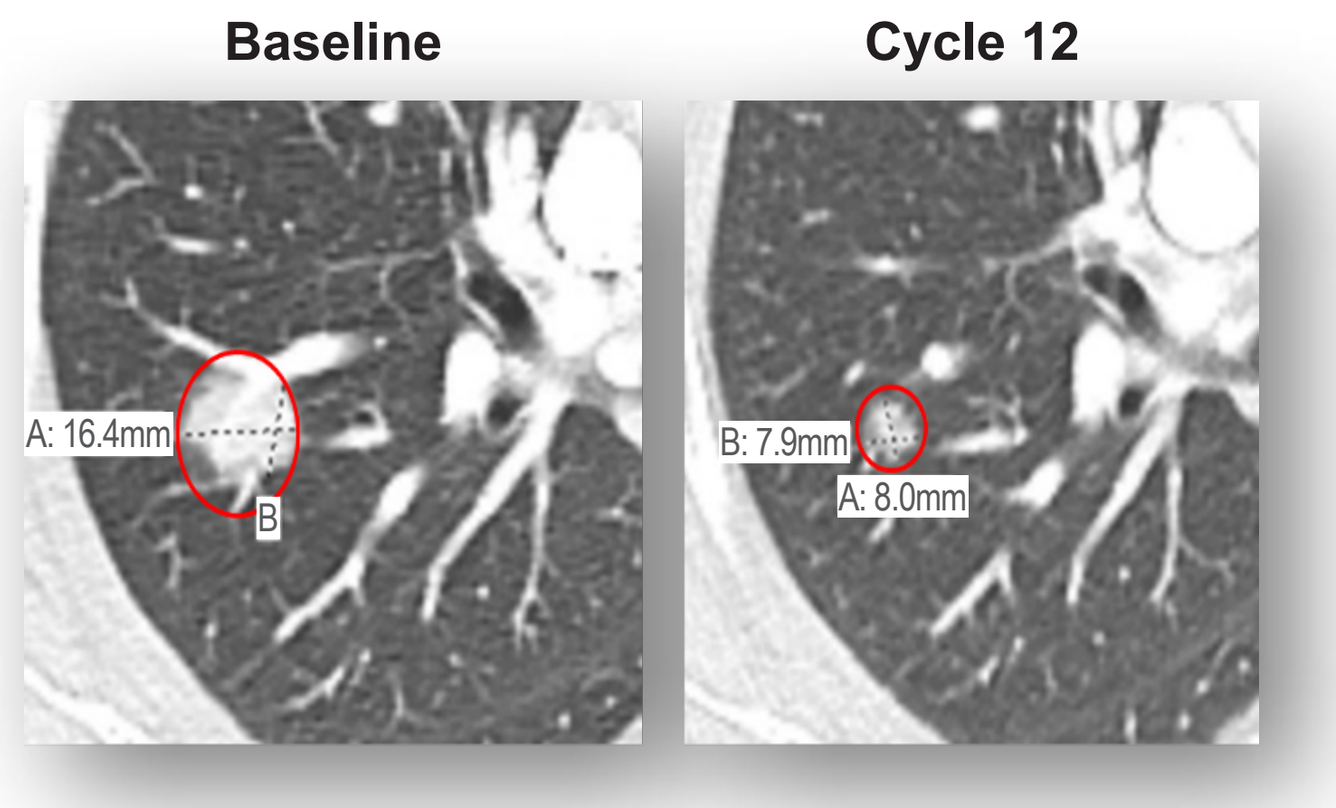
Baseline
Cycle 12
A: 16.4mm
B
B: 7.9mm
A: 8.0mm

CONCLUSION

- Tisotumab vedotin is an ADC that targets TF
- The recommended phase II dose of tisotumab vedotin is 2.0 mg/kg IV q3w
 - 3 DLTs were observed at the 2.2 mg/kg dose
- Tisotumab vedotin demonstrated a dose-dependent PK profile
- Tisotumab vedotin has demonstrated anti-tumour activity in this heavily pretreated patient population
- Patients enrolled in the expansion part of this study will provide further insight into the clinical activity of tisotumab vedotin in patients with advanced cancer of the cervix, ovary, endometrium, prostate, bladder, and oesophagus

REFERENCES

1. Breij EC et al. *Cancer Res*. 2014;74(4):1214-1226.
2. Chu AJ. *Int J Inflam*. 2011;2011. doi: 10.4061/2011/367284.
3. De Goeij BE et al. *Mol Cancer Ther*. 2015;14(5):1130-1140.
4. Clinical trials.gov. https://clinicaltrials.gov/ct2/show/NCT02001623. Accessed August 7, 2017.

ACKNOWLEDGEMENTS

This study was sponsored by Genmab A/S. Scientific writing was provided by Ogilvy CommonHealth, under the guidance of the primary author Dr. Chenard-Poirier, and funded by Genmab A/S.

Robert Coleman served on the scientific advisory committee for Genmab. Johann De Bono served as an advisory board member for Genmab, and his institution has received research funding from Genmab. David Hong has received research funding (for this trial) from Genmab. Steen Lisby is a former employee of Genmab.

Please contact the corresponding author for any questions: maxime.chenard-poirier@rmh.nhs.uk

Copies of this poster obtained through the quick response (QR) code are for personal use only and may not be reproduced without the written permission of the authors.

Scan this QR code with your electronic device to receive a PDF of this poster.

Poster presented at ESMO 2017 September 8-12, Madrid, Spain.